2236

Effectiveness of TBP-PI-HBr in Patients with Instrumentation, Anatomic or Functional Abnormalities: Secondary Analysis from ADAPT-PO

Angela K. Talley[1], Paul Eckburg[1], Ian Critchley[1], Gary Moore[2], Nivedita Bhatt[1], Lori Muir[1], David Melnick[1]

[1]Spero Therapeutics, Cambridge, MA; [2]Moore Computing Services, Inc., Little Rock, AR

SPERO THERAPEUTICS

Angela K. Talley, MD
Spero Therapeutics
675 Massachusetts Ave
14th Floor
Cambridge, MA 02139
E-mail: atalley@sperotherapeutics.com

Background

- There is an urgent need for new oral agents for the treatment of treatment of complicated urinary tract infections, including acute pyelonephritis (cUTI/AP); tebipenem pivoxil hydrobromide (TBP-PI-HBr) is a novel candidate oral carbapenem antibiotic in clinical development for the treatment of cUTI/AP.
- Published results of the Phase 3 ADAPT-PO trial[1] showed TBP-PI-HBr was non-inferior to IV ertapenem (ERT) in the treatment of patients with cUTI/AP as per the pre-specified analyses outlined for the study.
- cUTI patients with certain anatomical abnormalities, functional (metabolic or neurological) disorders, urinary tract instrumentation (i.e., indwelling catheters) and/or severe disease are at increased risk for poor outcomes, including recurrent bacteriuria and clinical relapse.

Objectives

- This secondary analysis of ADAPT-PO was undertaken to evaluate outcomes in patients with cUTI and/or AP and certain risk factors for recurrent bacteriuria and poor outcomes.

Methods

- ADAPT-PO was a global, Phase 3, randomized, double-blind, double-dummy noninferiority trial conducted between June 2019 and May 2020.[1] The primary objective was to evaluate the efficacy and safety of oral TBP-PI-HBr vs. IV ertapenem (ERT) in hospitalized adult patients with cUTI/AP.
- Hospitalized patients $\geq$18 years with a diagnosis of cUTI or AP were randomized 1:1 to receive oral TBP-PI-HBr 600mg every 8 hours (q8h), or IV ERT 1g, every 24 hours (q24h), for a total duration of 7-10 days, or up to 14 days in the setting of bacteremia at baseline, per investigator discretion. **(Figure 1)**
- The microbiological intent-to-treat population (micro-ITT) included all patients with a diagnosis of cUTI or AP and a positive urine culture at screening (growth of one or two uropathogens at $\geq 10^5$ colony-forming units [CFU] per mL, excluding certain pathogens not expected to respond to either study drug.
- The primary efficacy endpoint was the overall response (composite of clinical cure and microbiologic response) at the test-of-cure (TOC) visit in the micro-ITT population based on the following definitions:
 - Clinical cure: complete resolution or clinically significant improvement of baseline signs and symptoms of cUTI/AP and no new symptoms requiring no further antimicrobial therapy.
 - Microbiological response: reduction in the baseline uropathogen(s) to $<10^3$ CFU/mL in a post-baseline urine culture and a negative repeat blood culture if positive at baseline.
- In this ADAPT-PO subgroup analysis, outcomes were assessed among patients with risk factors for poor outcomes and/or recurrent bacteriuria, with focus on anatomic or functional abnormalities of the urinary tract.
- Risk factors were assessed by review of medical and surgical history to identify those with anatomical disorders or urinary tract instrumentation/procedures and/or functional (metabolic or neurological) disorders).
- Additional risk factors for poor outcomes included advanced age (≥65 years), disease severity (modified SIRS criteria at baseline), and bacteremia at baseline.

Figure 1. ADAPT-PO Trial Design

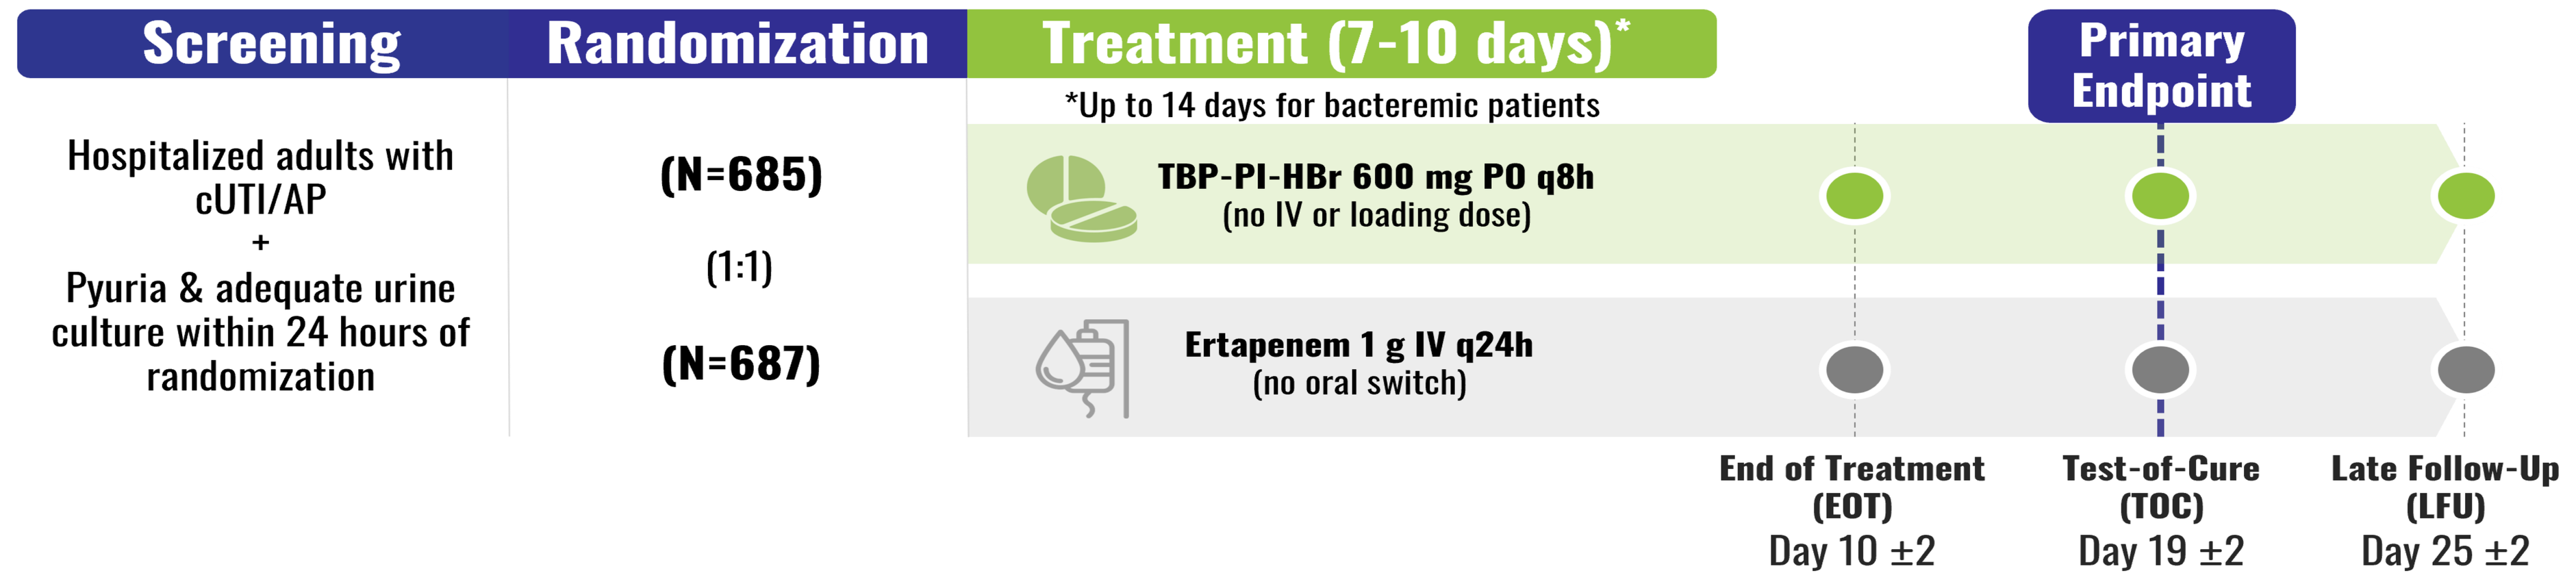
Screening
Randomization
Treatment (7-10 days)*
Primary Endpoint
*Up to 14 days for bacteremic patients
Hospitalized adults with cUTI/AP
+
Pyuria & adequate urine culture within 24 hours of randomization
(N=685)
(1:1)
(N=687)
TBP-PI-HBr 600 mg PO q8h
(no IV or loading dose)
Ertapenem 1 g IV q24h
(no oral switch)
End of Treatment (EOT) Day 10 ±2
Test-of-Cure (TOC) Day 19 ±2
Late Follow-Up (LFU) Day 25 ±2

Results

Table 1. Baseline characteristics and risk factors (micro-ITT)

	TBP-PI-HBr (N=449)	Ertapenem (N=419)
Age, years (mean ± SD)	**57.6 ± 18.7**	**58.7 ± 17.9**
Age >=65 years, n (%)	**203 (45.2)**	**197 (47.0)**
Female, n (%)	**252 (56.1)**	**253 (60.4)**
Baseline diagnosis, n (%)		
Acute pyelonephritis (AP)	**226 (50.3)**	**201 (48.0)**
cUTI	**223 (49.7)**	**218 (52.0)**
cUTI with AP	94/223 (42.2)	87/218 (39.9)
cUTI without AP	129/223 (57.8)	131/218 (60.1)
Bacteremia at baseline	**47 (10.5)**	**53 (12.6)**
Modified SIRS criteria, n (%)[a]	**98 (21.8)**	**73 (17.4)**
History of anatomical or functional abnormality, n (%)	**270 (60.1)**	**261 (62.3)**
Anatomical disorders or instrumentation, n (%)	**252 (56.1)**	**242 (57.8)**
Anatomical disorder, n (%)[b]	**243 (54.1)**	**232 (55.4)**
Benign prostatic hyperplasia	89/243 (54.1)	94/232 (40.5)
Nephrolithiasis	50/243 (36.6)	51/232 (22.0)
Urinary retention	42/243 (17.3)	28/232 (12.1)
Urethral stenosis	12/243 (4.9)	26/232 (11.2)
Urinary calculus	18/243 (7.4)	17/232 (7.3)
Chronic kidney disease	16/243 (6.6)	19/232 (8.2)
Renal cyst	15/243 (6.2)	19/232 (8.2)
Ureterolithiasis	17/243 (7.0)	14/232 (6.0)
Hydronephrosis	17/243 (7.0)	8/232 (3.4)
Neurogenic bladder	18/243 (7.4)	7/232 (3.0)
Functional (Metabolic or Neurological) disorders, n (%)[c]	**90 (20.0)**	**85 (20.3)**
Type 2 diabetes mellitus	54/90 (60.0)	46/85 (54.1)
Diabetes mellitus (unspecified)	27/90 (30.0)	25/85 (29.4)
Diabetic neuropathy	8/90 (8.9)	1/85 (1.2)
Paraplegia	3/90 (3.3)	2/85 (2.4)
Type 1 diabetes mellitus	3/90 (3.3)	2/85 (2.4)
Urinary tract instrumentation/procedure, n (%)[b]	**100 (22.3)**	**69 (16.5)**
Urethral bladder catheter	42/100 (42.0)	24/69 (34.8)
Bladder catheter	33/100 (33.0)	19/69 (27.5)
Stent	29/100 (29.0)	20/69 (29.0)
Suprapubic bladder catheter	16/100 (16.0)	15/69 (21.7)
Bladder catheter removal	11/100 (11.0)	10/69 (14.5)
Nephrostomy tube	8/100 (8.0)	6/69 (8.7)
Bladder catheter replacement	4/100 (4.0)	5/69 (7.2)
Uncomplicated AP with no risk factors[d], n (%)	**67 (14.9)**	**58 (13.8)**
≥ 1 risk factor[d], n (%)	**382 (85.1)**	**361 (86.2)**
≥ 2 risk factors[d], n (%)	**289 (64.4)**	**271 (64.7)**

[a] Modified criteria for systemic inflammatory response syndrome (SIRS)
[b] Only those representing >10% overall micro-ITT patients presented
[c] Only functional abnormalities representing >5% overall micro-ITT patients presented
[d] Risk factors for recurrent bacteriuria and/or poor outcome.

- In the overall study, 1372 patients were enrolled, and 868 (63.3%) were included in microbiological intent-to-treat population including 50.8% with cUTI and 49.2% with AP. **(Table 1)**
- At baseline, 382/449 (85.1%) and 361/449 (86.2%) of patients in the TBP-PI-HBr and ertapenem arms, respectively had ≥1 risk factor and 289 (64.4%) and 268 (64.2%) had ≥2 risk factors.
- The proportion of patients with various anatomical and or functional disorders were generally balanced between treatment groups: anatomical abnormalities and/or urinary tract instrumentation were present in 252 (56.1%) and 242 (57.8%) of patients in the TBP-PI-HBr and ertapenem arms, respectively; slightly more TBP-PI-HBr patients (22.3%) than ertapenem patients (16.5%) had recent history of urinary tract instrumentation. **(Table 1)**
- Among patients with ≥1 risk factor, overall response at TOC was 56.3% and 59.6% with TBP-PI-HBr and ERT, respectively. **(Table 2)**
- Overall response rates were higher in patients with no risk factors, achieved in 73.1% of subjects treated with TBP-PI-HBr and 74.1% among those treated with ertapenem.
- Up to LFU, overall response was lowest among patients with history of urinary tract instrumentation. **(Table 2)**
- Clinical response rates at TOC were high and similar in both arms in patients with (≥92.9%) and without (≥91.4%) risk factors. **(Table 2)** Across subsets of patients with Instrumentation, Anatomic or Functional Abnormalities, clinical response rates at TOC were high (≥87.8%) and generally balanced between treatment arms.
- At LFU, the rates of clinical relapse were low for both patients without risk factors (1 patient treated with ertapenem) and for patients with had ≥1 risk factors (3.1% and 3.9% for patients treated with TBP-PI-HBr and ertapenem, respectively). **(Table 2)**

Table 2. Clinical and overall response by visit in patients with and without recurrent bacteriuria risk factors (micro-ITT)

	Favorable Clinical Response[1]					
	EOT		TOC		LFU	
Patient Population	TBP-PI-HBr n/N (%)	Ertapenem n/N (%)	TBP-PI-HBr n/N (%)	Ertapenem n/N (%)	TBP-PI-HBr n/N (%)	Ertapenem n/N (%)
Anatomical Disorders	241/243 (99.2)	225/232 (97.0)	221/243 (90.9)	216/232 (93.1)	209/243 (86.0)	202/232 (87.1)
Functional (Metabolic/Neurological) Disorders	89/90 (98.9)	81/85 (95.3)	79/90 (87.8)	78/85 (91.8)	72/90 (80.0)	75/85 (88.2)
Urinary Tract Instrumentation/Procedures	100/100 (100.0)	64/69 (92.8)	91/100 (91.0)	61/69 (88.4)	88/100 (88.0)	56/69 (81.2)
Anatomical Disorders or Urinary Tract Instrumentation/Procedures	250/252 (99.2)	235/242 (97.1)	229/252 (90.9)	226/242 (93.4)	217/252 (86.1)	212/242 (87.6)
Anatomical or Functional (Metabolic or Neurological) Disorders	268/270 (99.3)	254/261 (97.3)	247/270 (91.5)	245/261 (93.9)	235/270 (87.0)	230/261 (88.1)
No Risk Factors for Recurrent Bacteriuria	67/67 (100.0)	56/58 (96.6)	63/67 (94.0)	53/58 (91.4)	62/67 (92.5)	54/58 (93.1)
≥ 1 RF for Recurrent Bacteriuria	379/382 (99.2)	354/361 (98.1)	355/382 (92.9)	339/361 (93.9)	336/382 (88.0)	323/361 (89.5)
≥ 2 RF for Recurrent Bacteriuria	287/289 (99.3)	262/269 (97.4)	265/289 (91.7)	253/269 (94.1)	250/289 (86.5)	239/269 (88.8)

	Favorable Overall Response					
	EOT		TOC		LFU	
Patient Population	TBP-PI-HBr n/N (%)	Ertapenem n/N (%)	TBP-PI-HBr n/N (%)	Ertapenem n/N (%)	TBP-PI-HBr n/N (%)	Ertapenem n/N (%)
Anatomical Disorders	234/243 (96.3)	217/232 (93.5)	126/243 (51.9)	124/232 (53.4)	113/243 (46.5)	107/232 (46.1)
Functional (Metabolic/Neurological) Disorders	86/90 (95.6)	76/85 (89.4)	37/90 (41.1)	39/85 (45.9)	31/90 (34.4)	30/85 (35.3)
Urinary Tract Instrumentation/Procedures	98/100 (98.0)	63/69 (91.3)	50/100 (50.0)	31/69 (44.9)	46/100 (46.0)	23/69 (33.3)
Anatomical Disorders or Urinary Tract Instrumentation/Procedures	243/252 (96.4)	227/242 (93.8)	131/252 (52.0)	127/242 (52.5)	118/252 (46.8)	110/242 (45.5)
Anatomical or Functional (Metabolic or Neurological) Disorders	261/270 (96.7)	243/261 (93.1)	140/270 (51.9)	142/261 (54.4)	126/270 (46.7)	122/261 (46.7)
No Risk Factors for Recurrent Bacteriuria	66/67 (98.5)	56/58 (96.6)	49/67 (73.1)	43/58 (74.1)	47/67 (70.1)	39/58 (67.2)
≥ 1 RF for Recurrent Bacteriuria	371/382 (97.1)	340/361 (94.2)	215/382 (56.3)	215/361 (59.6)	199/382 (52.1)	191/361 (52.9)
≥ 2 RF for Recurrent Bacteriuria	280/289 (96.9)	253/269 (94.1)	154/289 (53.3)	150/269 (55.8)	141/289 (48.8)	129/269 (48.0)

[1] Clinical response includes clinical cure (EOT, TOC) or sustained clinical cure (LFU)
EOT: end of treatment; LFU: late follow up; TOC: test of cure; RF: risk factor for recurrent bacteriuria and/or poor outcomes

Summary and Conclusions

- As expected based on the definition of cUTI, the majority of patients in ADAPT-PO had anatomical or functional GU abnormalities and/or recent history of urinary tract instrumentation or procedures, placing them at high risk for recurrent bacteriuria after the cessation of antibiotic treatment.
- Among cUTI patients with ≥1 baseline risk factor for recurrent bacteriuria and/or poor outcome, response rates were comparable between oral tebipenem pivoxil hydrobromide and IV ertapenem. When assessed per the composite primary efficacy outcome of combined clinical and microbiological response, overall response rates were higher at TOC and LFU visits in both groups among patients without these risk factors.
- Clinical cure rates among these at-risk patient subgroups were high at TOC and generally maintained in both treatment groups up to LFU.
- The high clinical cure rates in both treatment groups across all visits (EOT, TOC, and LFU) suggest that most patients with unfavorable overall response at TOC and LFU had asymptomatic bacteriuria, which would not typically require treatment in the clinical setting.
- These data provide additional supportive evidence of the clinical efficacy of oral TBP-PI-HBr for the treatment of patients with cUTI/AP.

References

1. Eckburg PB, Muir L, Critchley IA, Walpole S, Kwak H, Phelan AM, Moore G, Jain A, Keutzer T, Dane A, Melnick D, Talley AK. Oral Tebipenem Pivoxil Hydrobromide in Complicated Urinary Tract Infection. *N Engl J Med.* 2022 Apr 7;386(14):1327-1338.

IDWeek 2022, October 19-23, Washington DC