Brian D. VanScoy, B.S.
Institute for Clinical Pharmacodynamics, Inc.
242 Broadway
Schenectady, NY 12305
Telephone: (518) 631-8100
Fax: (518) 631-8199
E-mail: BVanscoy@ICPD.com

Oral Tebipenem as Step-Down Therapy Following Intravenous Ertapenem in a 7-day Hollow-Fiber *In Vitro* Infection Model

B.D. VanScoy[1], S. Jones[1], H. Conde[1], L.V. Friedrich[2], N. Cotroneo[2], P.G. Ambrose[1]
[1]Institute for Clinical Pharmacodynamics, Inc., Schenectady, NY; [2]Spero Therapeutics, Cambridge, MA

INTRODUCTION

- Intravenous (IV) to oral (PO) antibiotic step-down therapy has many benefits, including reducing the length of a hospital stay and lowering the risk of nosocomial infections and overall cost [1, 2, 3].
- While fluoroquinolone agents are utilized as a step-down therapy for the treatment of patients with urinary tract infections, safety concerns [4] and increases in fluoroquinolone-resistant *Escherichia coli* [5, 6] makes this class of agents an increasingly unsuitable option.
- Tebipenem is an orally bioavailable carbapenem, which is administered as a pro-drug (tebipenem pivoxil hydrobromide) with broad-spectrum activity against ESBL positive pathogens, currently in development for the treatment of patients with complicated urinary tract infections (cUTI).

OBJECTIVE

- The objective of these studies was to use 7-day hollow-fiber *in vitro* infection model assays designed to evaluate the potential of PO tebipenem as step-down therapy from ertapenem IV therapy.

METHODS

Antimicrobial Agent and Challenge Isolates

- A panel of five *Escherichia coli* isolates were selected based on their known resistance mechanisms and tebipenem minimum inhibitory concentration (MIC) values. All isolates were either purchased from JMI Laboratories (North Liberty, IA) or provided by the National Collection of Type Cultures (NCTC) (**Table 1**).
- Tebipenem was provided by Spero Therapeutics (Cambridge, MA). Ertapenem was purchased from Henry Schein (Melville, NY).

Table 1. Known resistance mechanisms and tebipenem and ertapenem modal MIC values of isolates utilized in the hollow-fiber *in vitro* infection model step-down studies

Isolate	Known resistance mechanisms	Modal MIC (mg/L)	
		Tebipenem	Ertapenem
E. coli NCTC 13441	CTX-M-15, Sequence Type-131 (ST-131)	0.015	0.03
E. coli 4643	CTX-M-15, OXA1/30	0.008	0.015
E. coli 1033345	CMY-2, TEM-1 (ST-131, O25b)	0.06	0.25
E. coli 998822	CTX-M-15, OXA-1, OXA-30 (ST-131 O25b)	0.03	0.015
E. coli 13319	CTX-M-15	0.015	0.125

Hollow-Fiber *In Vitro* Infection Model

- 10 mL of each *E. coli* isolate were inoculated into the hollow-fiber *in vitro* infection model cartridges (Fibercell Systems, Frederick, MD) at an inoculum of 1.0 x 10^7 colony forming units (CFU)/mL using Mueller-Hinton broth medium (**Figure 1**).
- Each challenge isolate was subjected to concentration-time profiles simulating free-drug plasma concentrations observed after tebipenem PO administration every eight hours (q8h) in healthy volunteers [7] or ertapenem IV administration every 24 hours (q24h)[8].
 - Free-drug concentration time profiles simulated assuming plasma protein binding values of 45 and 90% for tebipenem and ertapenem, respectively.

METHODS

- Each isolate was challenged in duplicate by an assortment of active and inactive regimens including:
 - Ertapenem administered as a 1 g IV dose q24h for 1, 3, or 7 days of therapy.
 - Tebipenem 600 mg PO q8h was simulated as a step-down therapy, following either 1 or 3 days of ertapenem treatment, as well as a 7-day monotherapy regimen.
 - Inactive control regimens including no-treatment, as well as ertapenem 1 g IV q24h for 1 or 3 days, followed by a halting of therapy.
- Samples were collected for enumeration of bacterial populations and observation of simulated PK profiles over the duration of the 7-day study period.

Figure 1. Schematic of the hollow-fiber *in vitro* infection model

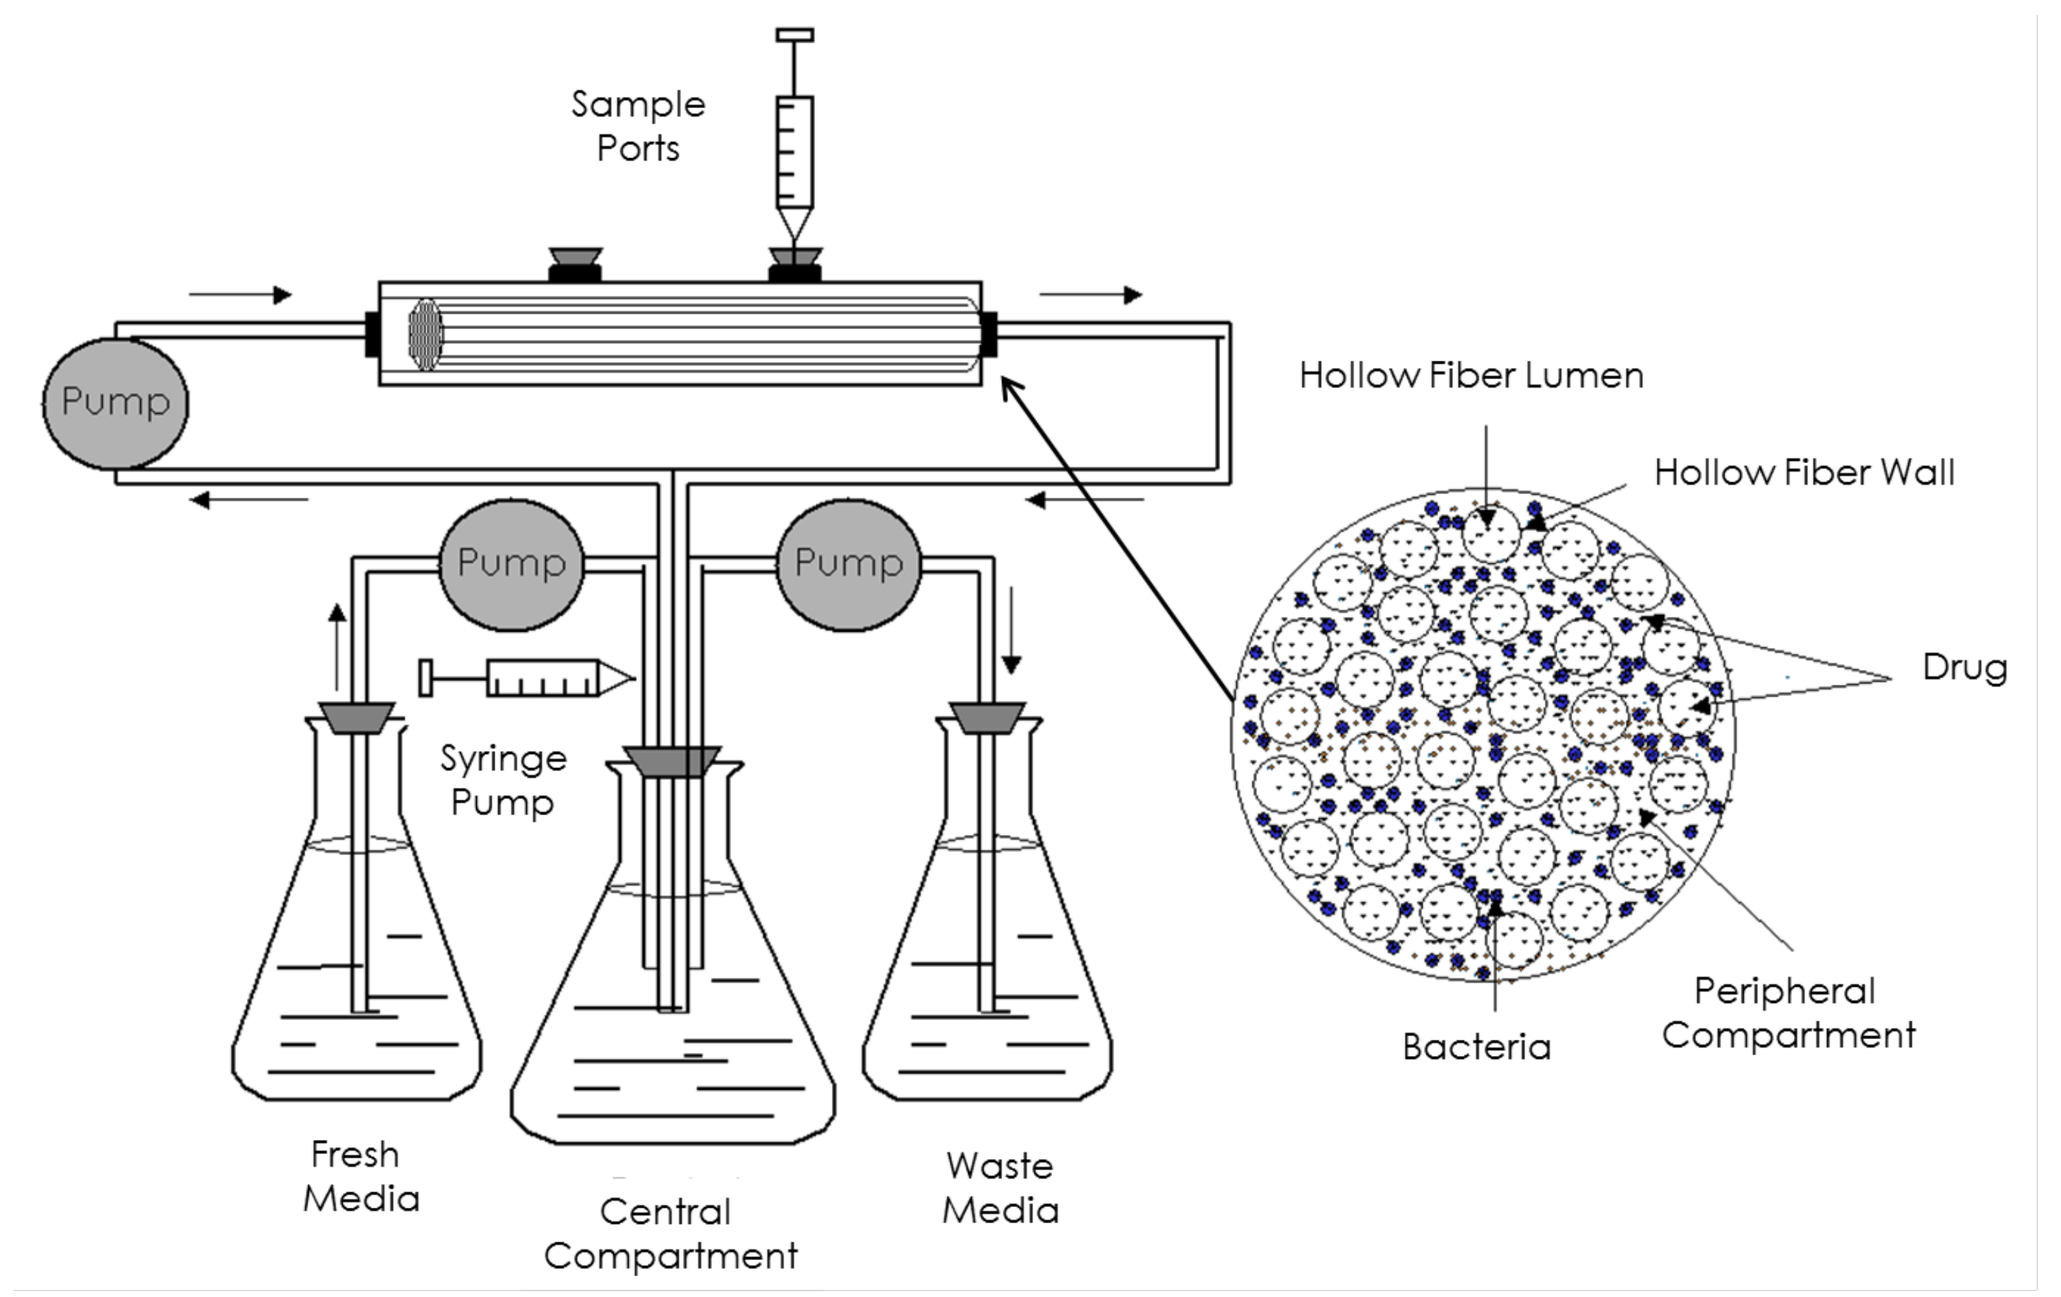
Sample Ports
Hollow Fiber Lumen
Hollow Fiber Wall
Drug
Pump
Pump
Pump
Syringe Pump
Bacteria
Peripheral Compartment
Fresh Media
Central Compartment
Waste Media

RESULTS

- As shown in **Figure 2**, good agreement was observed between the targeted and observed concentration-time profiles for tebipenem and ertapenem simulated in the hollow fiber *in vitro infection* model.
- An example of the observed concentration-time profiles simulated in the hollow fiber *in vitro infection* model for each of the dosing regimens evaluated over the 7-day period are provided in **Figure 3.**

Figure 2. The relationship between targeted and observed tebipenem and ertapenem concentrations simulated in the hollow-fiber *in vitro* infection models

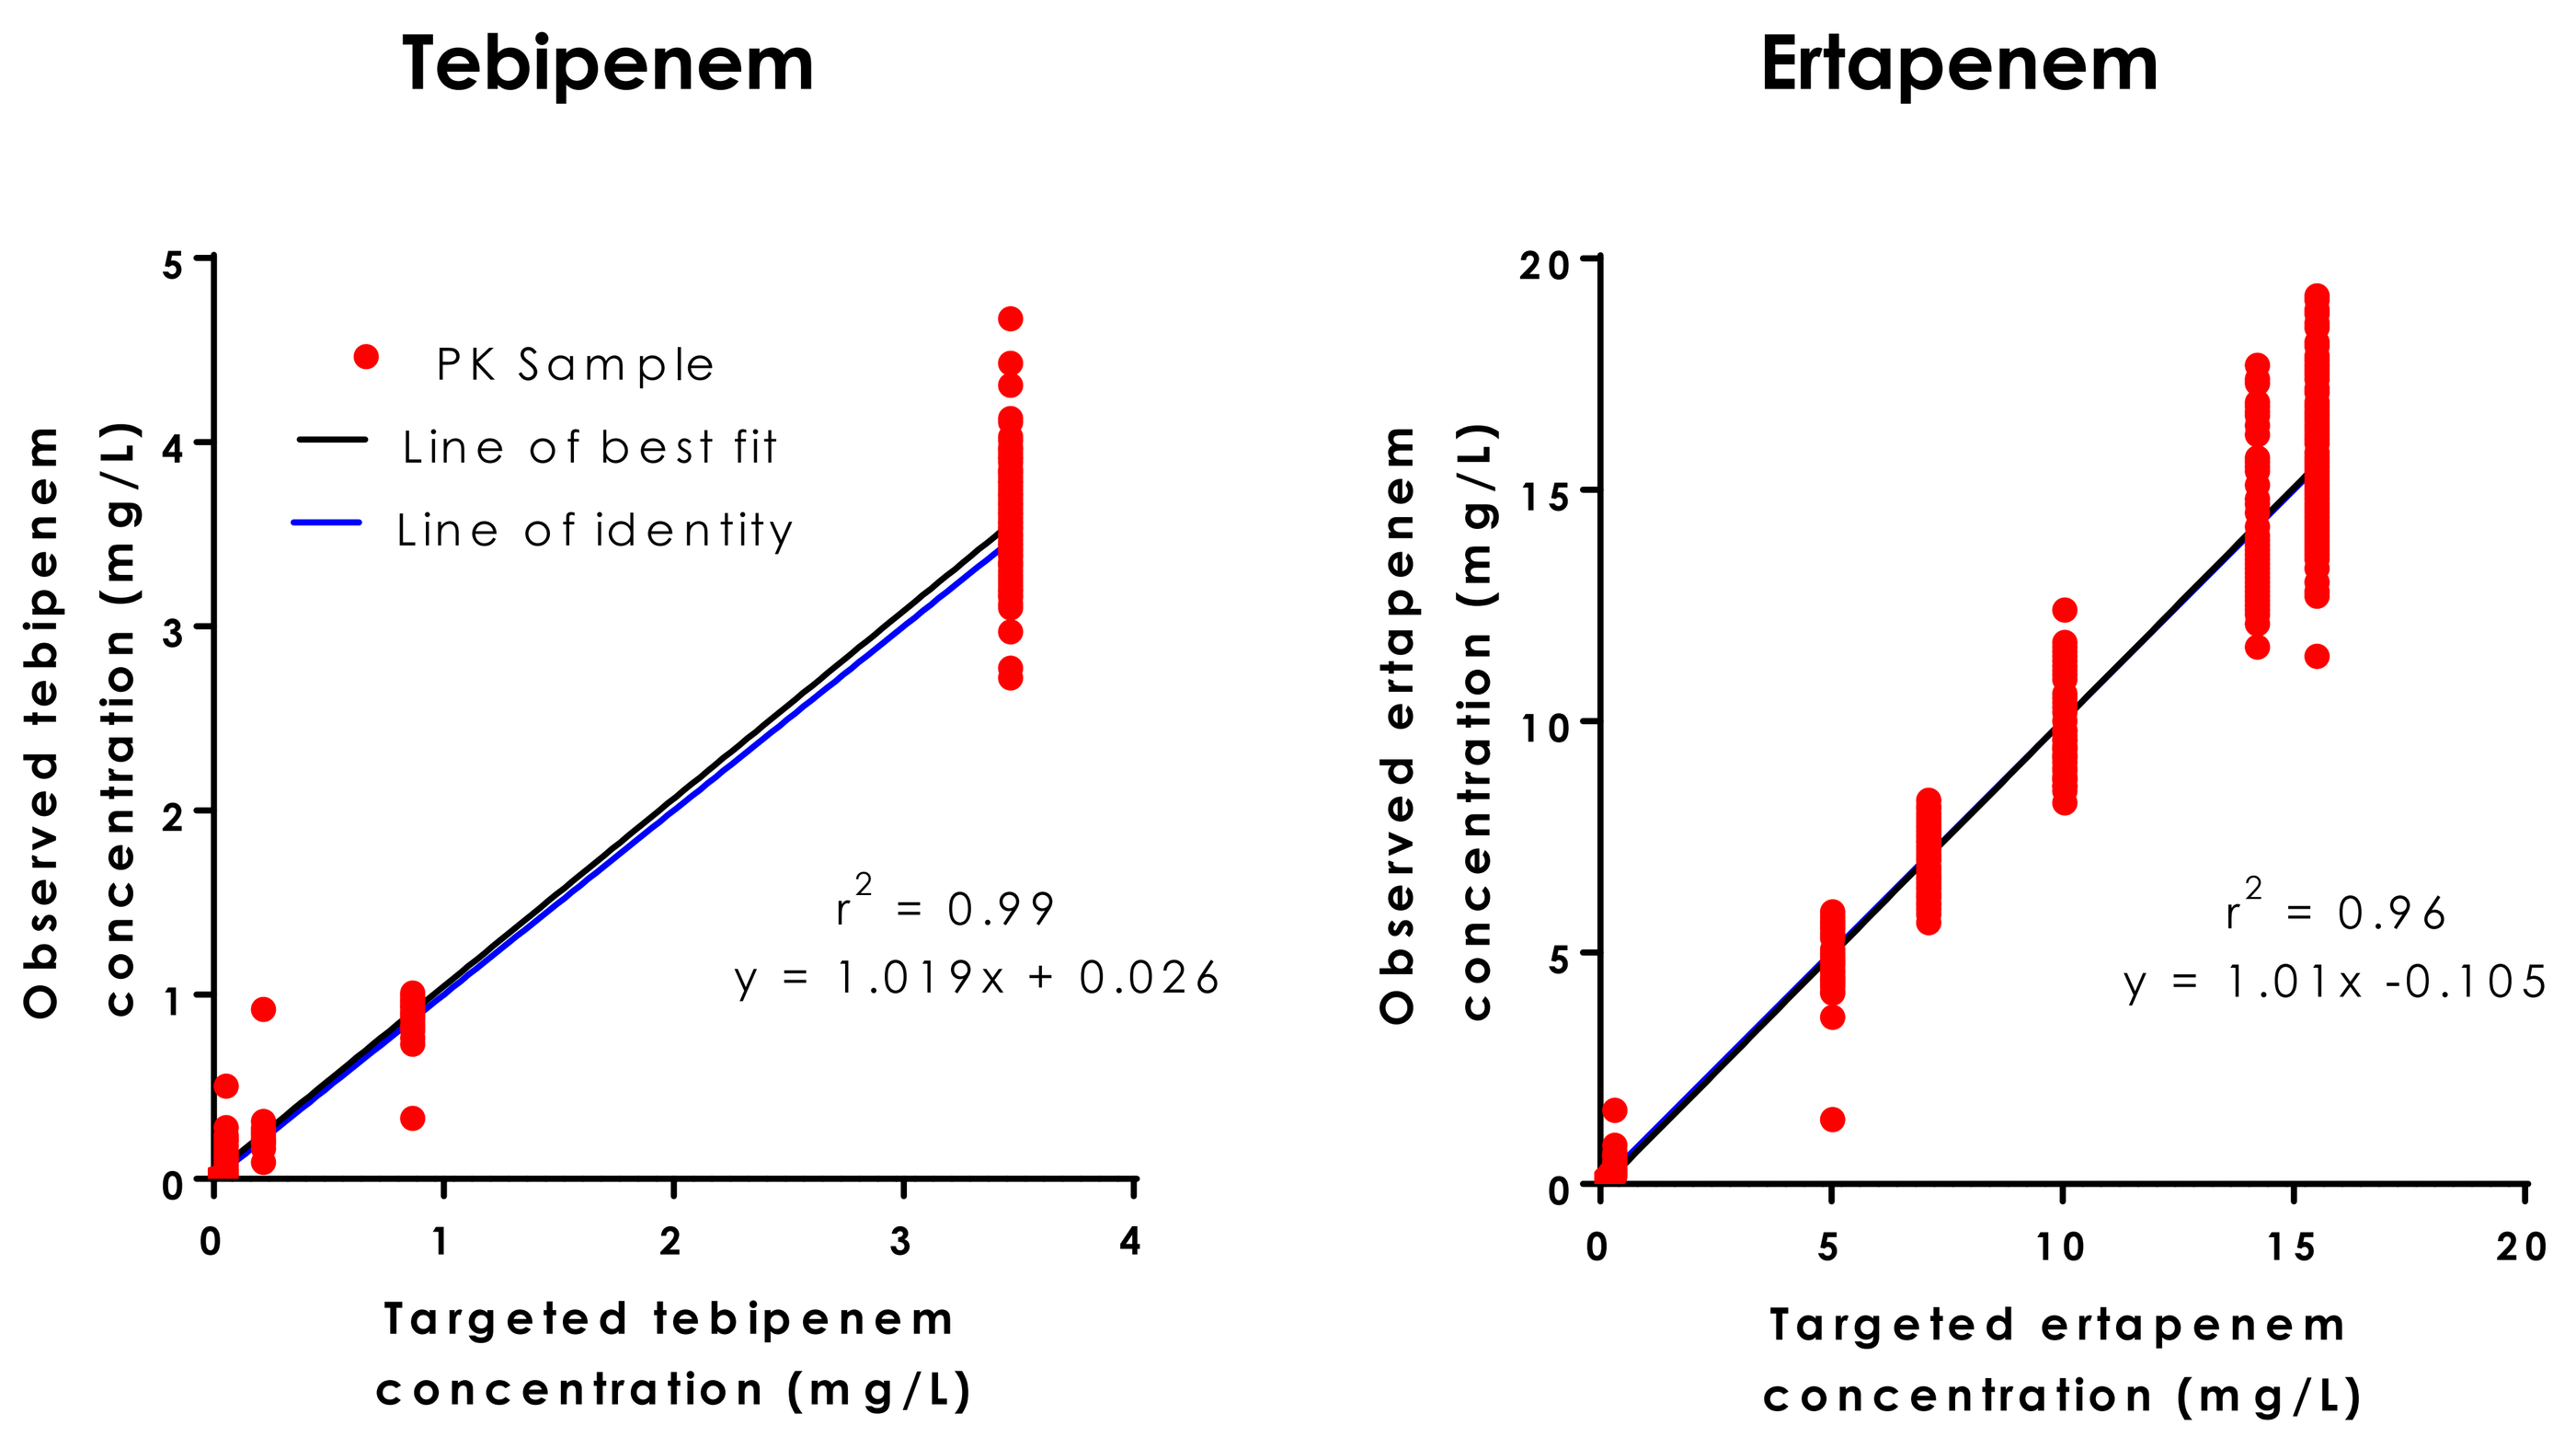
Tebipenem
PK Sample
Line of best fit
Line of identity
r² = 0.99
y = 1.019x + 0.026
Observed tebipenem concentration (mg/L)
Targeted tebipenem concentration (mg/L)
Ertapenem
r² = 0.96
y = 1.01x -0.105
Observed ertapenem concentration (mg/L)
Targeted ertapenem concentration (mg/L)

RESULTS

- The average results of the 7-day hollow fiber *in vitro* infection model step-down studies, displayed as bacterial burden over time for each dosing regimen evaluated, are displayed in **Figure 4**.
- Bacteria grew well in the no-treatment control group, reaching densities >10^{10} CFU/mL for all five isolates.
- The tebipenem 600 mg PO q8h monotherapy dosing regimen successfully reduced bacterial burdens below that initially inoculated in the system for four out of five isolates evaluated.
- The ertapenem monotherapy dosing regimens successfully reduced the bacterial burden to densities lower than that of the initial burden, as well as below limit of detection, for all five isolates evaluated over the 7-day period.
- Immediate regrowth, matching that observed in the no-treatment control was observed in the hollow-fiber *in vitro infection* model when ertapenem treatment was halted following both one and three days of treatment.
- The tebipenem 600 mg PO q8h dosing regimen successfully prevented bacterial regrowth following either one or three days of ertapenem IV therapy for all five *E. coli* isolates evaluated.

Figure 3. Targeted concentration time profiles for tebipenem and ertapenem in the hollow-fiber *in vitro* infection model, with observed data overlaid for a sample of evaluated regimens

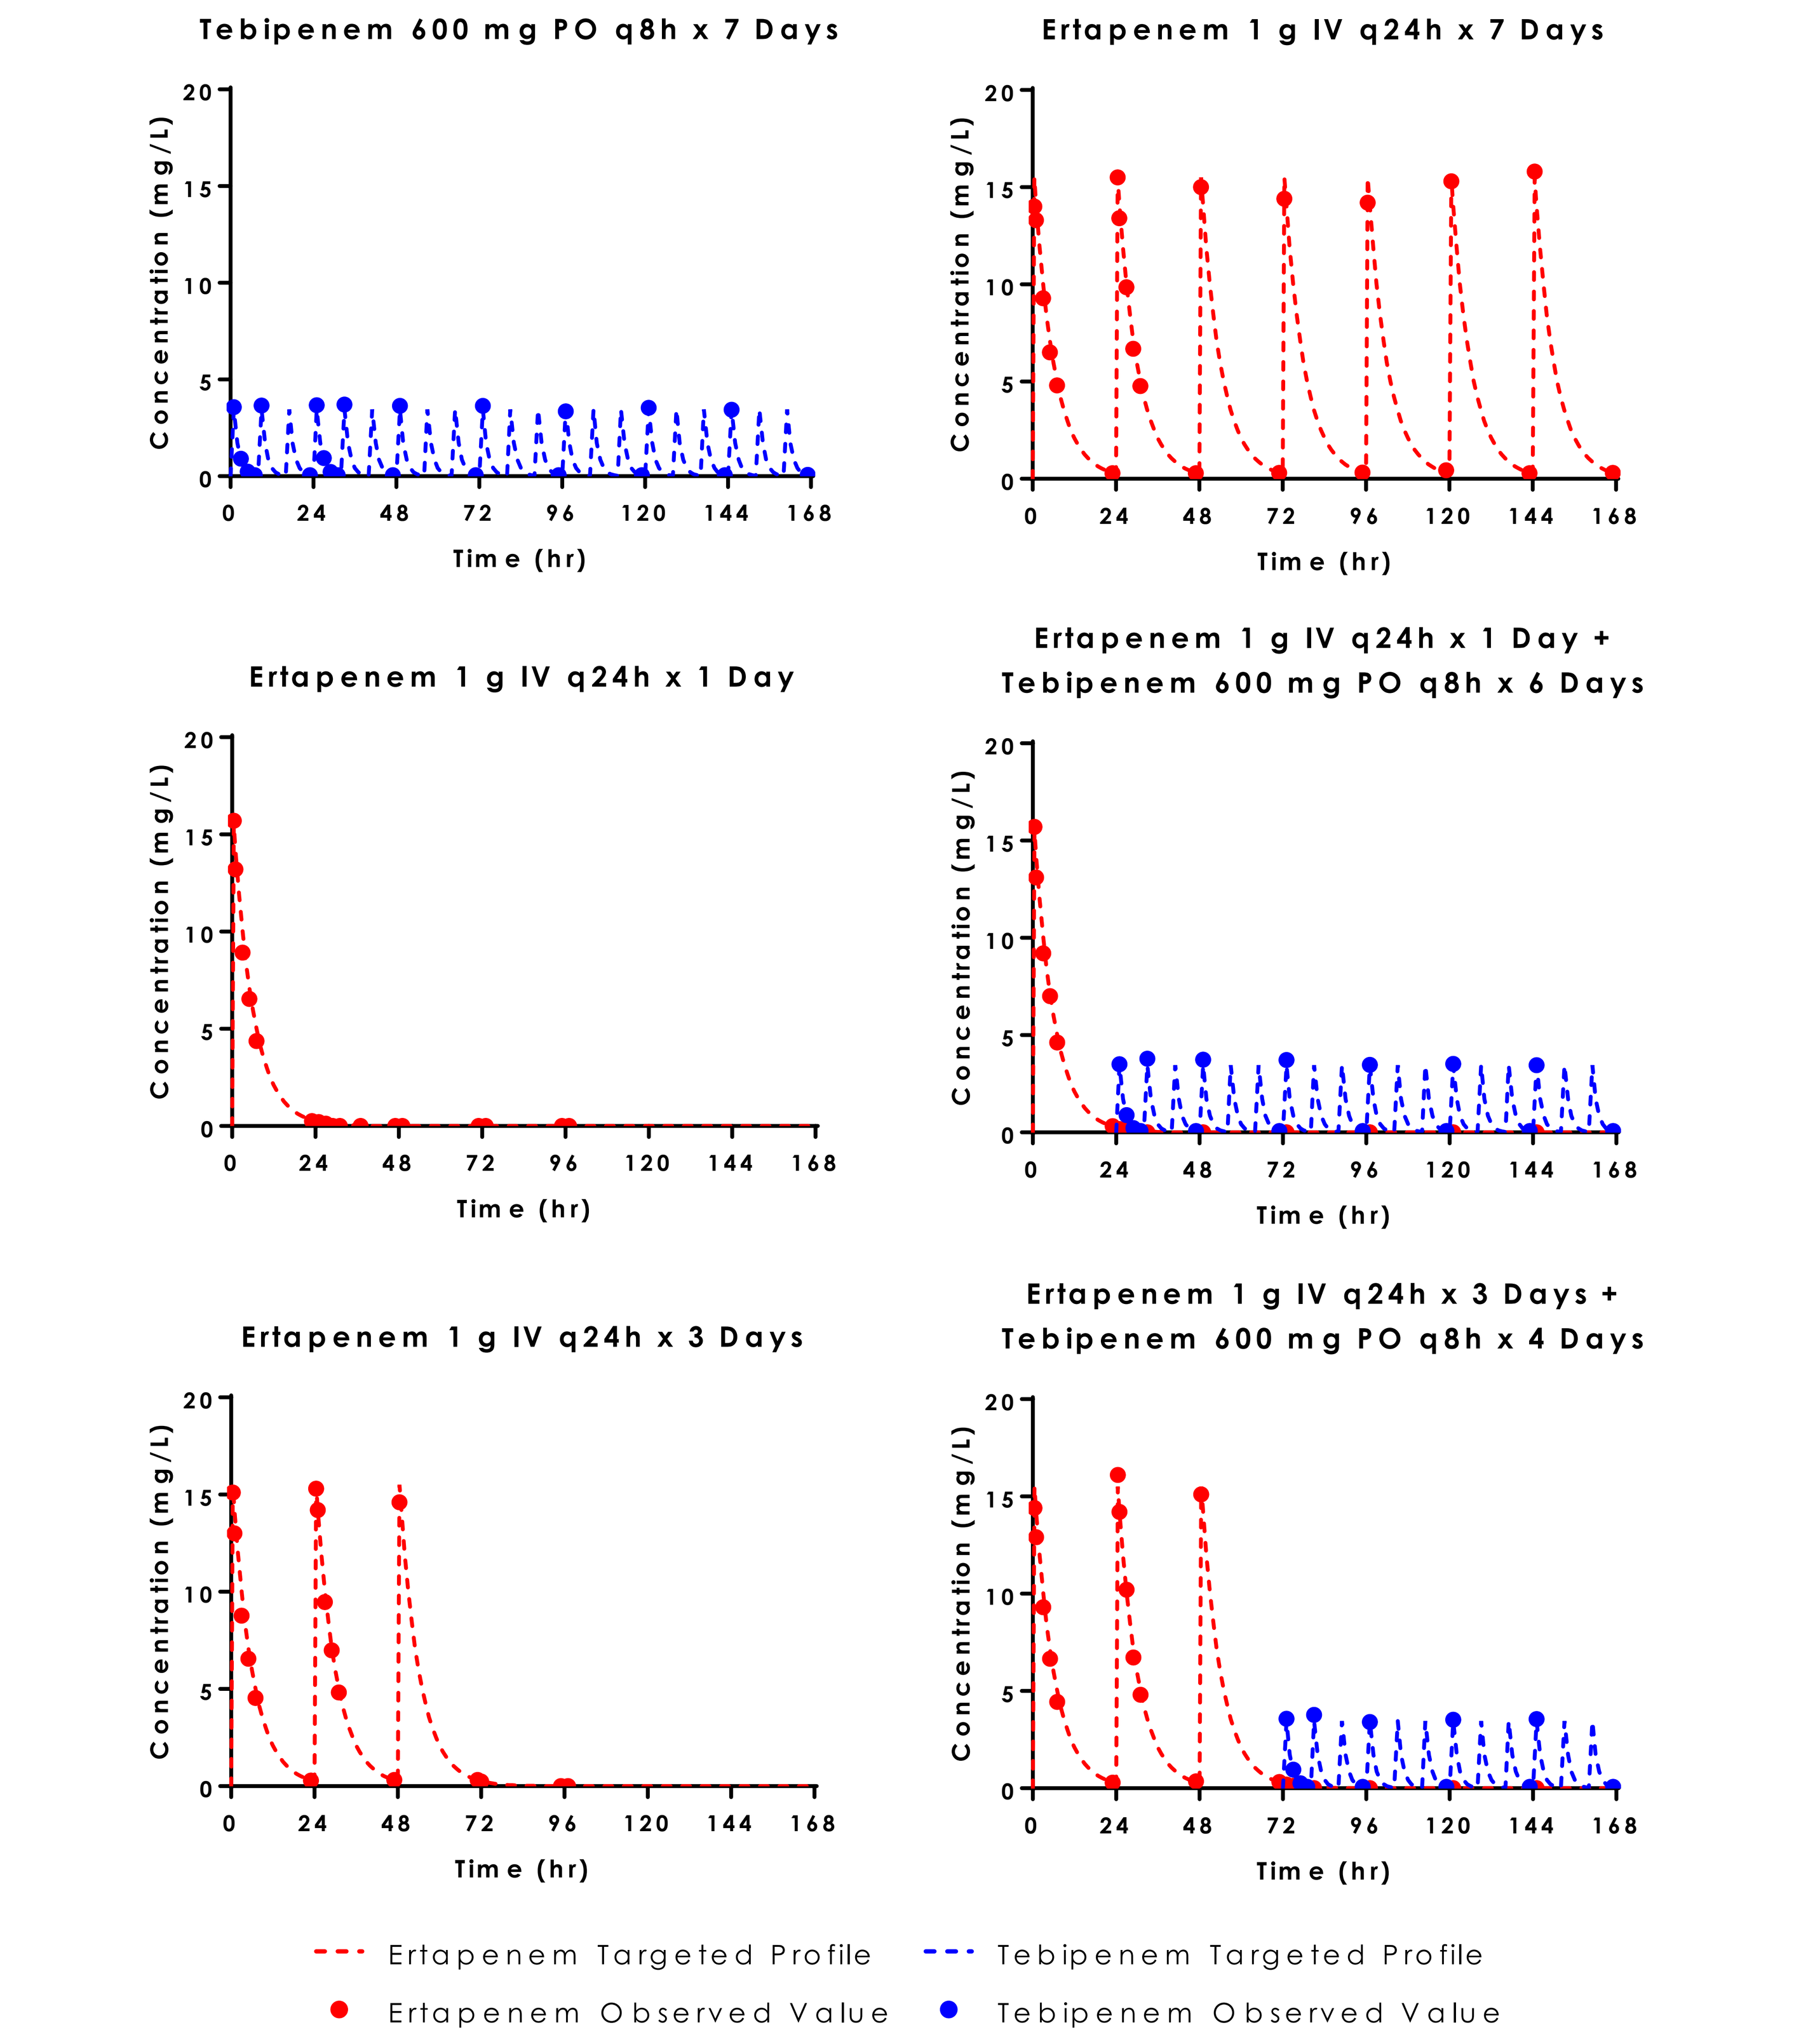
Tebipenem 600 mg PO q8h x 7 Days
Ertapenem 1 g IV q24h x 7 Days
Ertapenem 1 g IV q24h x 1 Day
Ertapenem 1 g IV q24h x 1 Day + Tebipenem 600 mg PO q8h x 6 Days
Ertapenem 1 g IV q24h x 3 Days
Ertapenem 1 g IV q24h x 3 Days + Tebipenem 600 mg PO q8h x 4 Days
Concentration (mg/L)
Time (hr)
Ertapenem Targeted Profile
Tebipenem Targeted Profile
Ertapenem Observed Value
Tebipenem Observed Value

RESULTS

Figure 4. Average bacterial burdens observed for five *E. coli* isolates in the 7-day hollow-fiber infection model evaluating ertapenem 1 g IV q24h and tebipenem 600 mg PO q8h dosing regimens as monotherapy and as step-down from IV to PO therapy

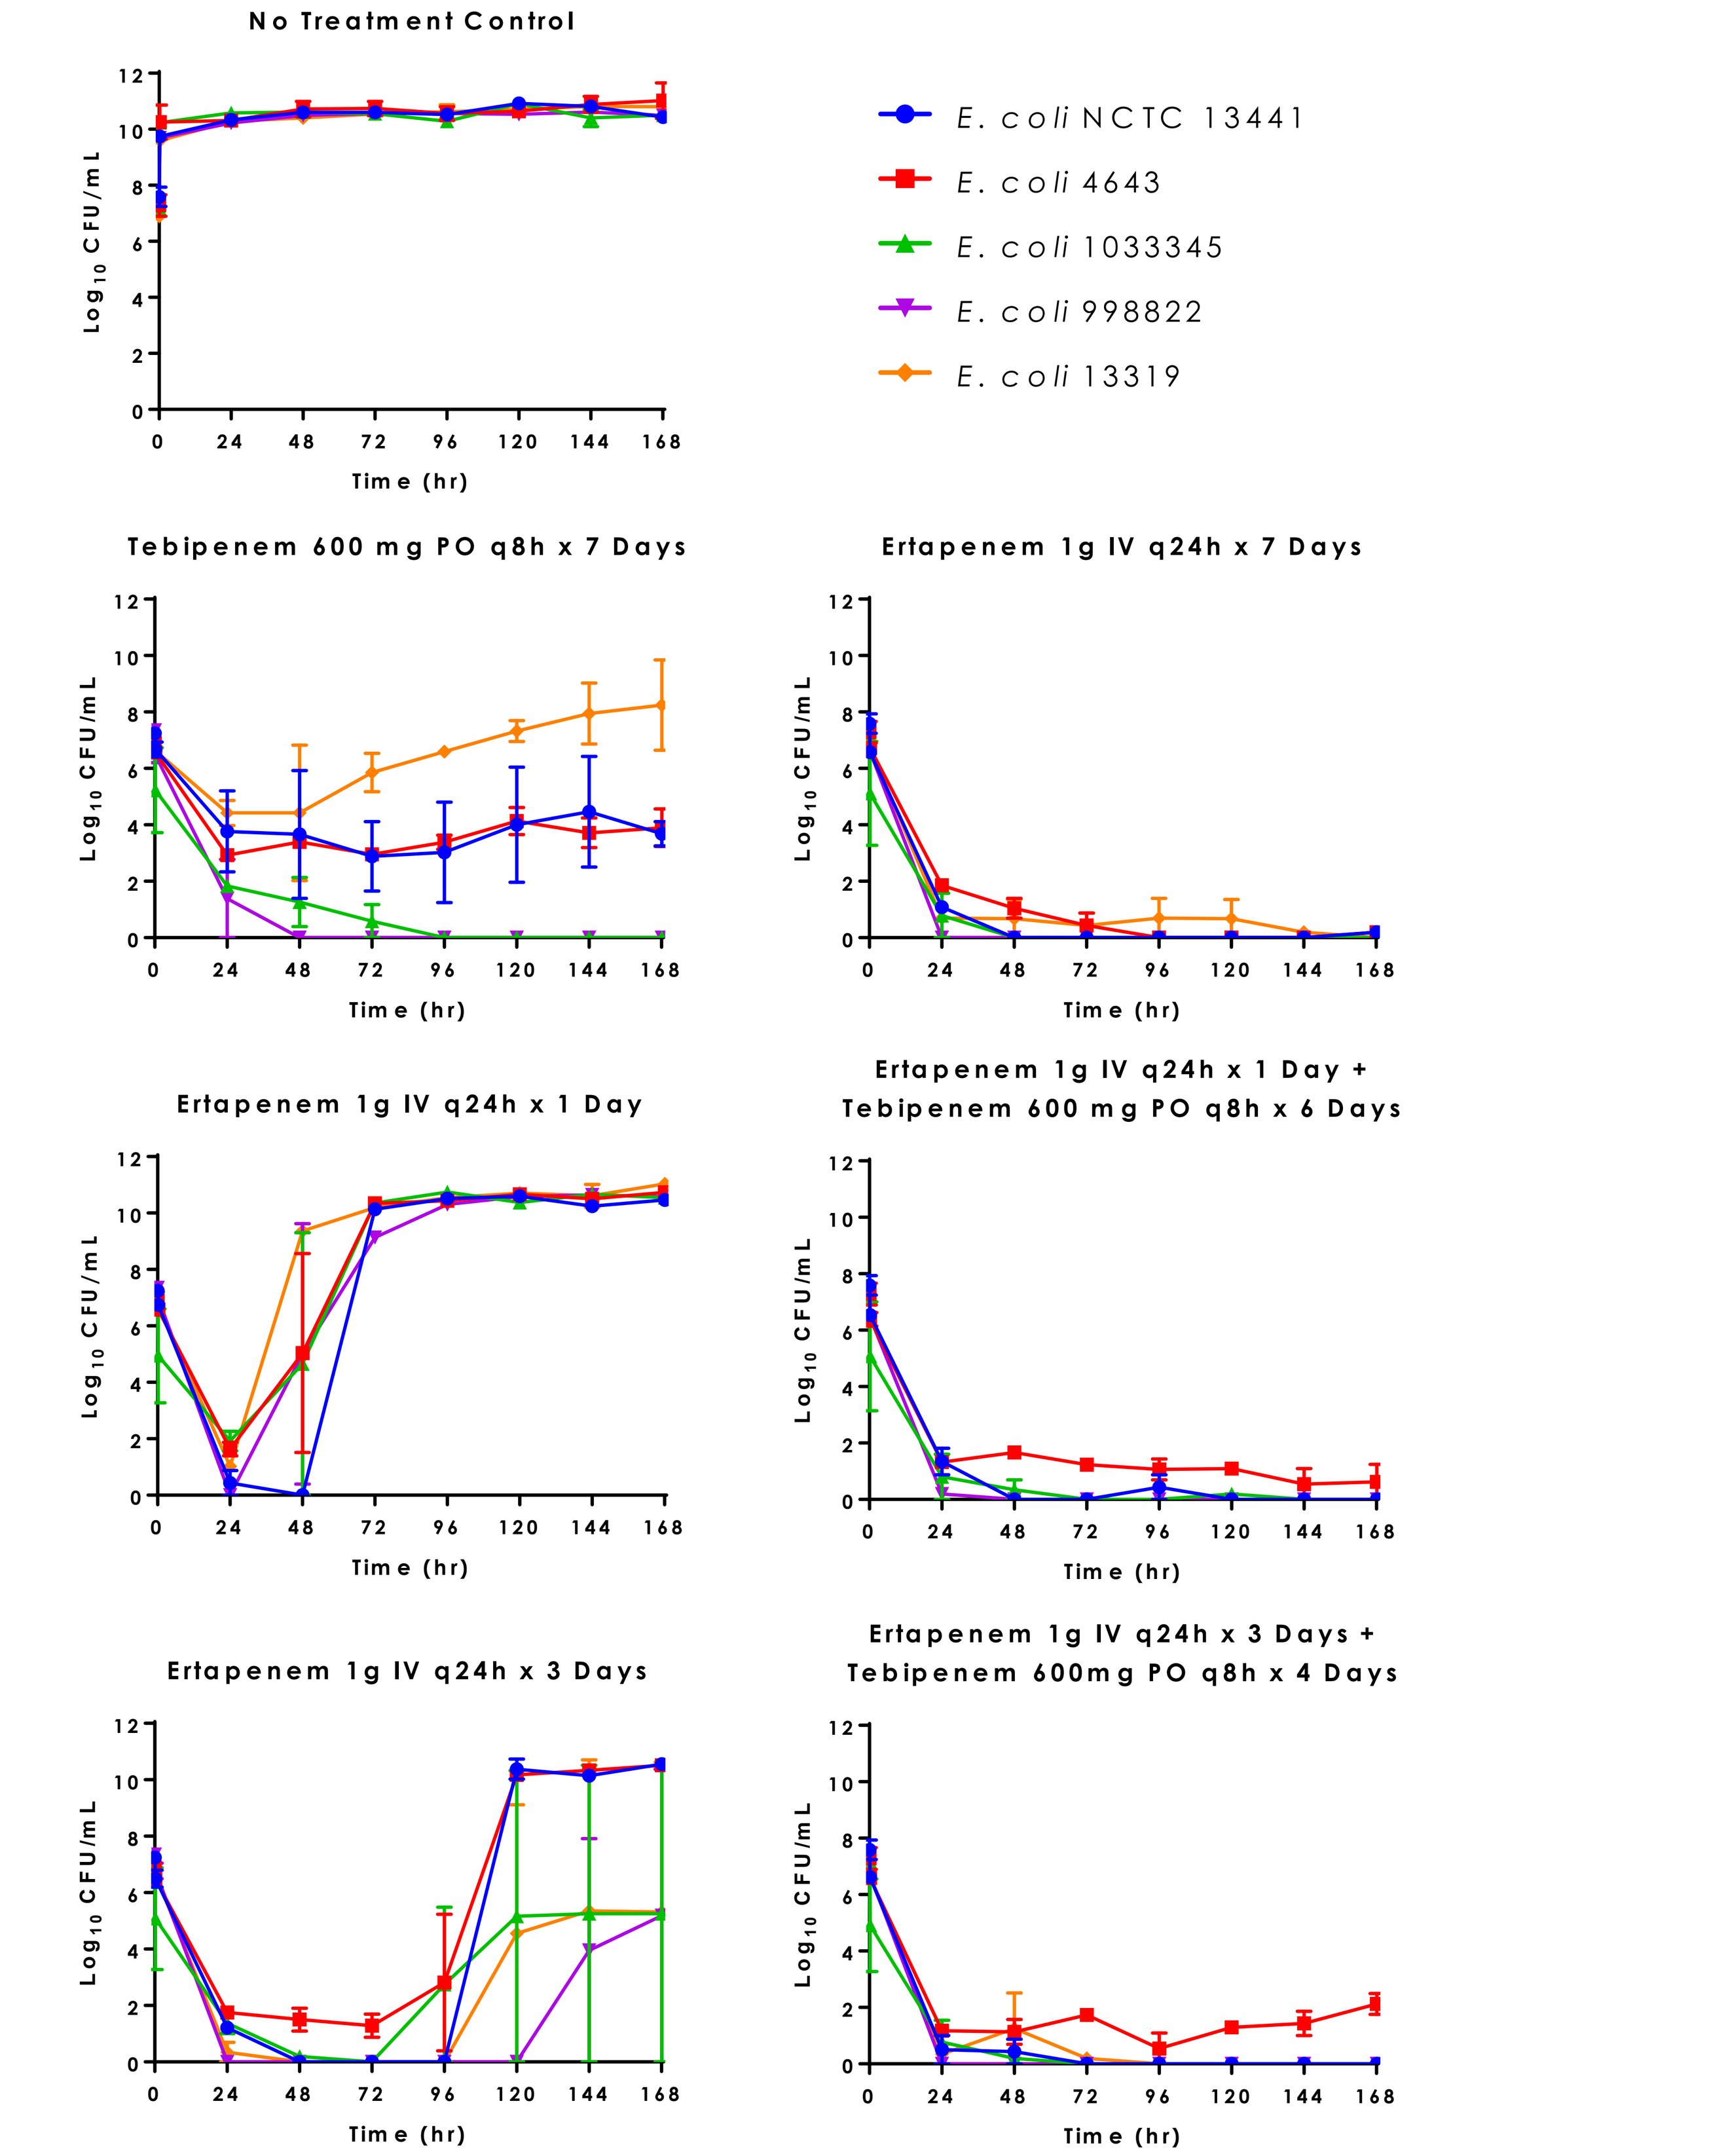
No Treatment Control
E. coli NCTC 13441
E. coli 4643
E. coli 1033345
E. coli 998822
E. coli 13319
Tebipenem 600 mg PO q8h x 7 Days
Ertapenem 1g IV q24h x 7 Days
Ertapenem 1g IV q24h x 1 Day
Ertapenem 1g IV q24h x 1 Day + Tebipenem 600 mg PO q8h x 6 Days
Ertapenem 1g IV q24h x 3 Days
Ertapenem 1g IV q24h x 3 Days + Tebipenem 600mg PO q8h x 4 Days
Log10 CFU/mL
Time (hr)

CONCLUSIONS

- These data demonstrate the potential utility of tebipenem as an PO step-down from ertapenem IV therapy for the treatment of cUTI arising from *E. coli*, including those producing ESBL. These data also suggest the need to evaluate tebipenem as a PO step-down after administration of other IV regimens.

REFERENCES

1. Béïque L, Zvonar R. Addressing concerns about changing the route of antimicrobial administration from intravenous to oral in adult inpatients. Can J Hosp Pharm 2015;68(4):318-326.
2. Sze WT, Kong MC. Impact of printed antimicrobial stewardship recommendations on early intravenous to oral antibiotics switch practice in district hospitals. Pharm Pract 2018;16(2):855.
3. Nathwani D, Lawson W, Dryden M, Stephens J, Corman S, Solem C, Li J, Charbonneau C, Baillon-Plot N, Haiser S, Eckmann C. Implementing criteria-based early switch/early discharge programmes: a European perspective. Clin Microbiol Infect 2015;21:S47-S55.
4. FDA Briefing Document; Joint Meeting of the Antimicrobial Drugs Advisory Committee and the Drug Safety and Risk Management Advisory Committee. The benefits and risks of systemic fluoroquinolone antibacterial drugs for the treatment of Acute Bacterial Sinusitis (ABS), Acute Bacterial Exacerbation of Chronic Bronchitis in Patients Who Have Chronic Obstructive Pulmonary Disease (ABECB-COPD), and Uncomplicated Urinary Tract Infections (uUTI). 2015; p.1–42.
5. Critchley IA, Cotroneo N, Pucci MJ, Mendes R. The burden of antimicrobial resistance among urinary tract isolates of *Escherichia coli* in the United States in 2017. PLoS One. 2019;14:e0220265.
6. Critchley IA, Cotroneo N, Pucci MJ, Jain A, Mendes RE. Resistance among urinary tract pathogens collected in Europe during 2018. J Glob Antimicrob Resist. 2020;23:439-444.
7. Data on file, Spero Therapeutics.
8. Invanz [package insert]. Whitehouse Station, NJ: Merck and Co., Inc.; 2021.

Poster no. 1113
IDWeek 2021, Virtual Conference, September 28-October 2, 2021