Pharmacodynamics of Ertapenem

[1]L. McEntee*, [1]S. Das, [1]A. Johnson, [1]N. Farrington, [1]S. Gore, [1]A. Kirby, [2]D. Melnick, [2]T. Parr, [1]W. Hope

[1]Antimicrobial Pharmacodynamics and Therapeutics, University of Liverpool, UK

[2]Spero Therapeutics, Cambridge, MA, USA

Laura McEntee, 15th April 2019, Poster number 7676

University of Liverpool, +44 (0) 151 794 5471, l.mcentee@liv.ac.uk

ABSTRACT

Background: Ertapenem is a licensed broad-spectrum carbapenem used to treat nosocomial infections. Little is published about the PK-PD of ertapenem, although it is expected that time-based indices (i.e. T>MIC) describe its PK-PD, as with other carbapenems. These studies suggest that ertapenem can be used as a benchmark for the development of novel compounds

Materials/methods: The pharmacokinetics of ertapenem were determined using a neutropenic mouse thigh infection model against a well-characterized wild-type strain of *Escherichia coli* ATCC 25922. Groups of 3 mice were dosed intravenously at doses of 8.33, 16.67, 33.33, 66.67mg/kg ertapenem every 8 hours for 24 hours. Mouse plasma was obtained in the 1st and 3rd dosing intervals, and plasma drug concentrations were measured via LC/MS/MS. The pharmacodynamics of ertapenem were investigated in the same mouse model with *E. coli* ATCC 25922, and 2 other ESBL-producing strains of *E. coli*. Groups of 3 mice were dosed intravenously at doses of ertapenem ranging from 0.25 - 66.67mg/kg. The endpoint was the average bacterial density in both thighs (CFU/g). The PD of ertapenem was described using both fT>MIC and fAUC:MIC*1/tau (another measure of time-dependent antimicrobial activity)

Results: Both indices of fT>MIC and fAUC:MIC*1/tau described the data effectively. An AUC:MIC*1/tau value of 48 and 60% fT>MIC achieved stasis in the mouse. MIC90 values for the *E. coli* strains studied ranged from 0.08 - 0.125mg/L

Conclusions: The PD of ertapenem was described equally well using both indices of fT>MIC and fAUC:MIC*1/tau, and using either index resulted in the same human dose prediction. As ertapenem can be described using more than one index, it is an important compound to benchmark against the development of novel compounds where there is little known about the PK-PD

INTRODUCTION

- Ertapenem is a licensed broad-spectrum carbapenem that is used to treat nosocomial infections
- There is currently minimal published PK-PD information available
- A PK-PD index of T>MIC is assumed for efficacy of ertapenem, as with other antibiotics of its class; the carbapenems
- The pharmacodynamics of ertapenem were explored in neutropenic mouse thigh models of infection
- It is suggested that ertapenem can be used as a benchmark for the development of novel compounds
- Both T>MIC and AUC:MIC*1/tau were used to described ertapenem

METHODS

- Neutropenic thigh CD-1 male mouse models conducted under a UK Home Office project license and approved by the Animal Welfare Ethics Review Board at the University of Liverpool
- Pharmacodynamics explored in a wild-type strain and 2 ESBL-producing strains of *E. coli*
- Endpoint of studies was average CFU/g of both thighs or drug concentrations (ug/L) in plasma

Dose Finding Studies

- Treatment was initiated 2 hours post inoculation. Ertapenem was administered intravenously within a dose range of 0.75 to 200mg/kg every 8 hours (q8h), against 3 different strains of *E. coli*
- An inhibitory Sigmoid Emax model was fitted to the data using ADAPT5

Pharmacokinetics (PK)

- The PK of ertapenem was estimated using a range of dosages; 8.33, 16.67, 33.33, and 66.67mg/kg, administered q8h intravenously
- Drug concentrations in mouse plasma were measured using LC/MS/MS in the 1st and 3rd dosing intervals
- Results were mathematically modelled using Pmetrics

Human Drug Exposures

- Monte Carlo simulations were used to calculate predicted human drug exposures
- A population PK model from Goutelle et al[1] was used
- Simulated patients received 1gram daily intravenously

RESULTS

1. Strains, molecular information, MICs

Organism	Strain Identifier	Molecular Information	Ertapenem MIC (ug/mL)
E. coli	ATCC 25922	QC strain	0.0078
	SPT 719	CP_SHV_ESBL,CP_TEM_WT	0.125
	SPT 731	CP_CTX-M_Group1,CP_TEM_WT, ST131 O25b clade	0.06

2. Pharmacokinetics of ertapenem

Figure 1 – Average murine plasma drug concentrations (ug/L, n=3)

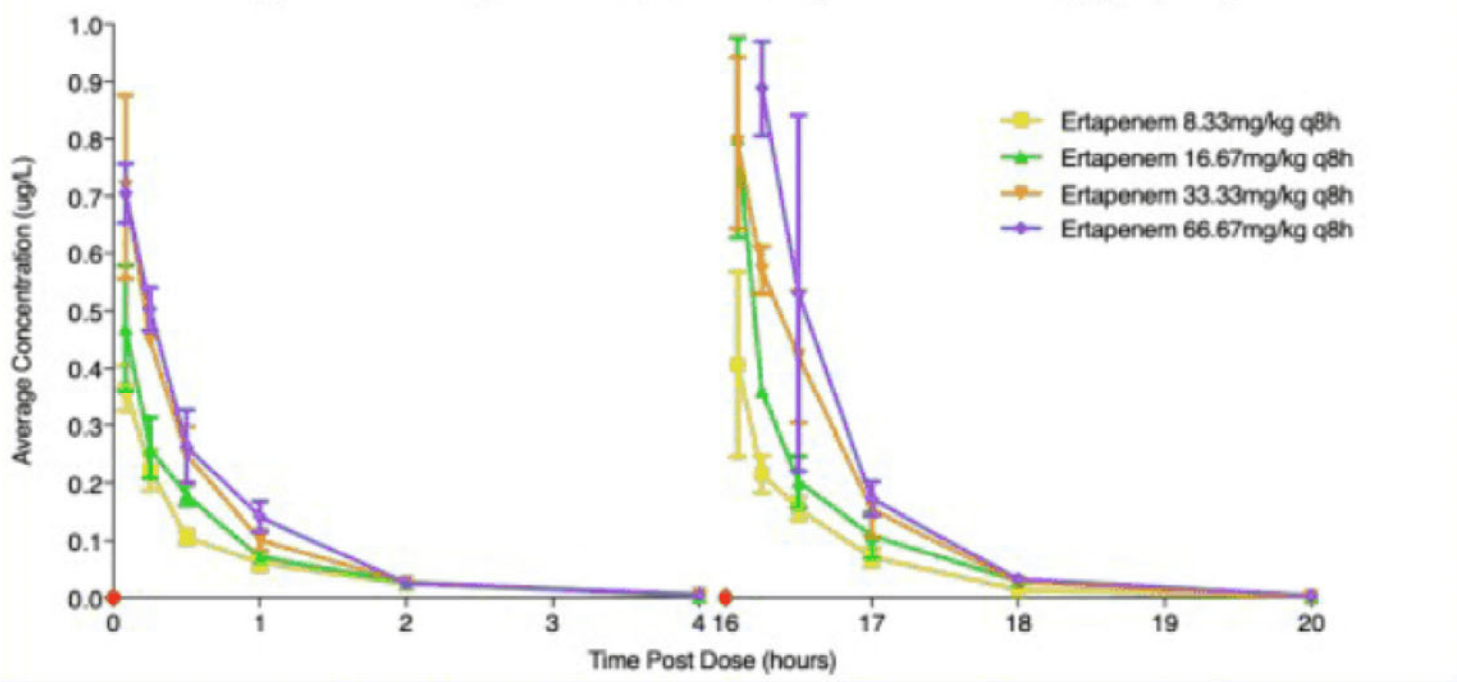
Average Concentration (ug/L)
Time Post Dose (hours)
Ertapenem 8.33mg/kg q8h
Ertapenem 16.67mg/kg q8h
Ertapenem 33.33mg/kg q8h
Ertapenem 66.67mg/kg q8h

3. Dose finding studies with 3 strains of *E. coli*. Two indices were used to describe the time dependency of ertapenem; fT>MIC (fig. 2) and fAUC:MIC*1/tau (fig. 3). Both indices equally described the data effectively. An fAUC:MIC*1/tau value of 48 and 60% fT>MIC achieved stasis in the mouse across all 3 strains of *E. coli*

Figure 2 – fT>MIC of 3 *E. coli* strains

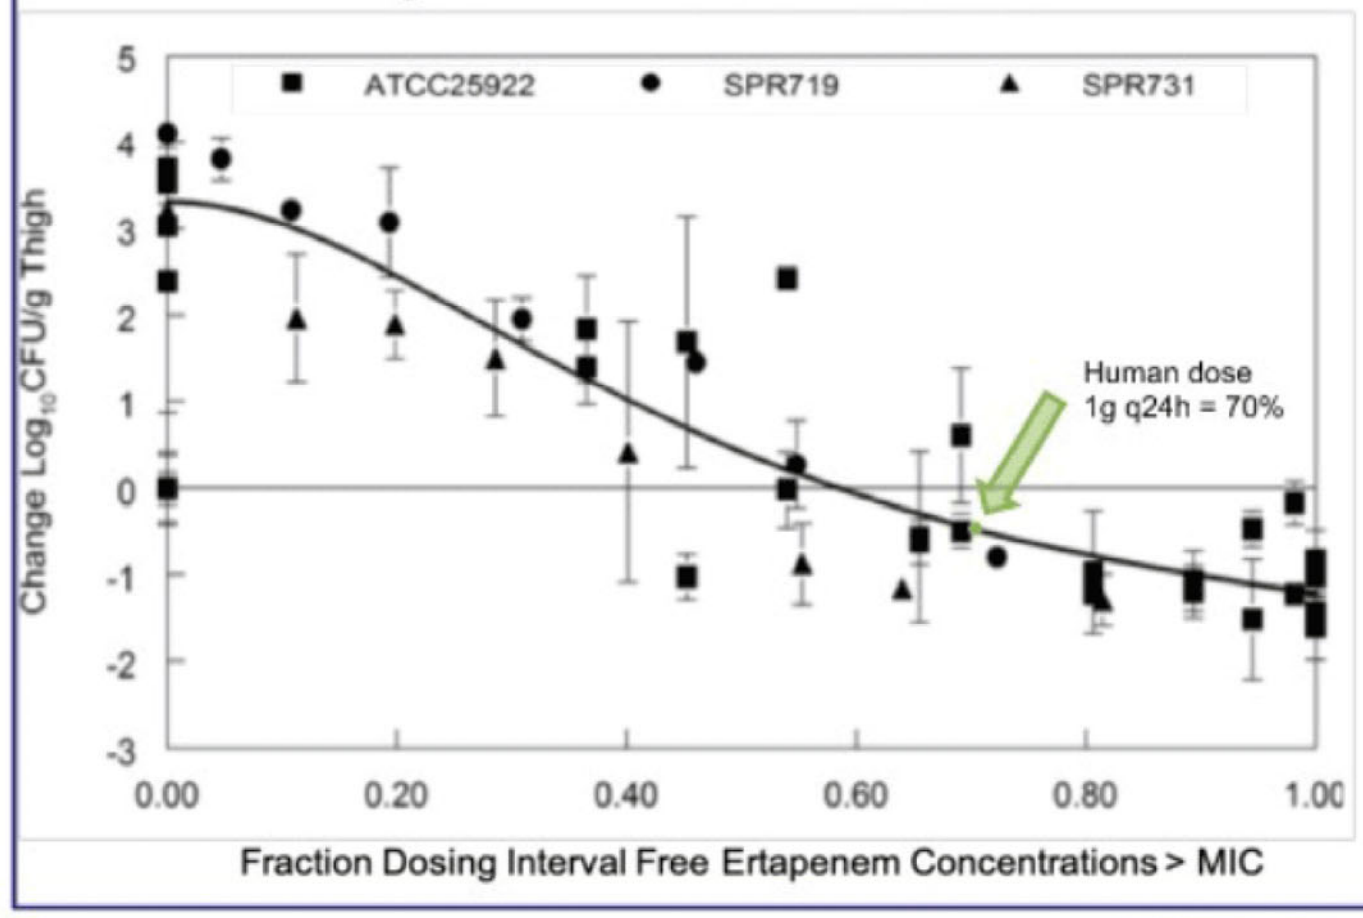
ATCC25922
SPR719
SPR731
Human dose
1g q24h = 70%
Change Log10CFU/g Thigh
Fraction Dosing Interval Free Ertapenem Concentrations > MIC

Figure 3 – fAUC:MIC*1/tau of 3 *E. coli* strains

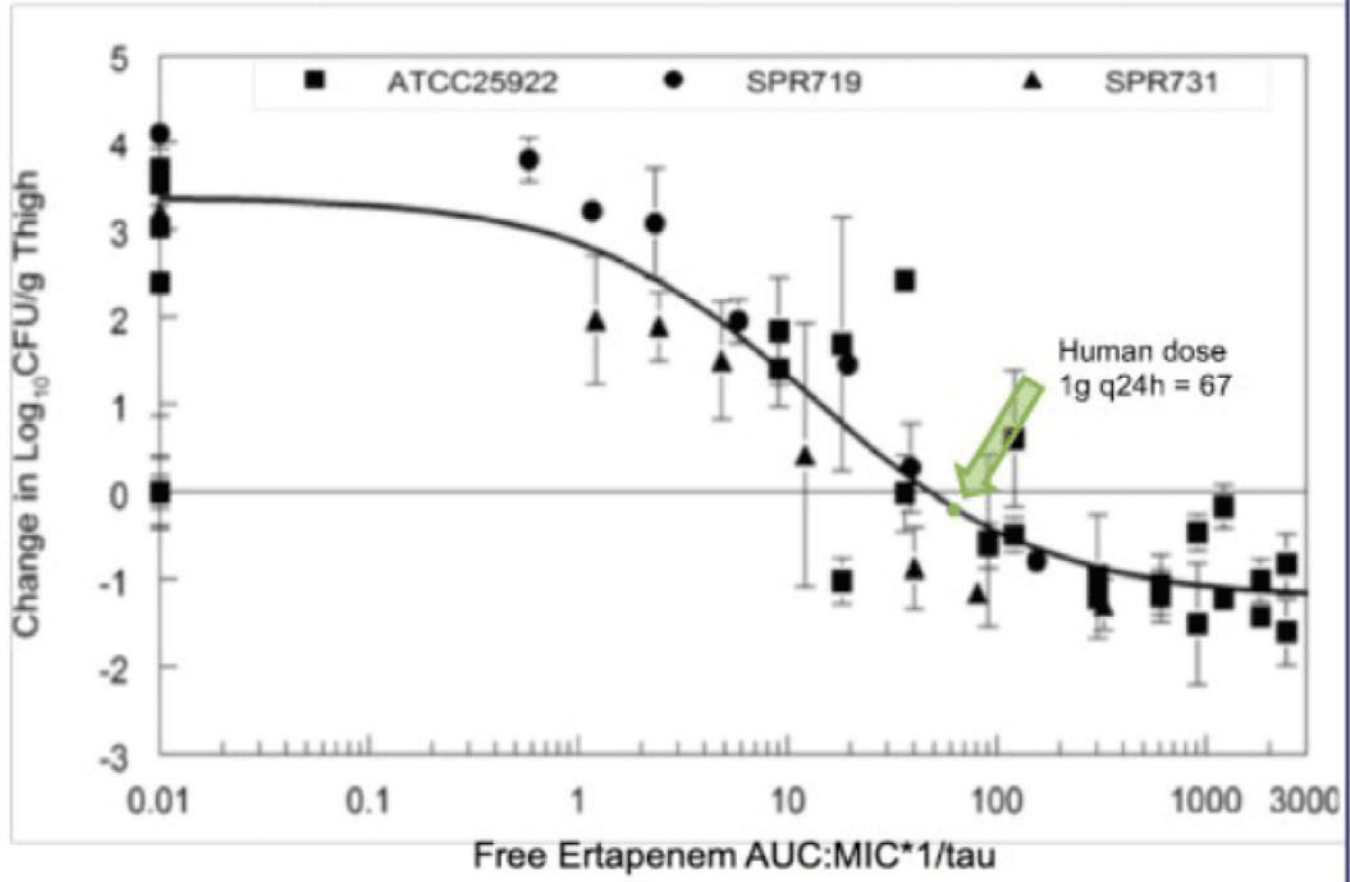
ATCC25922
SPR719
SPR731
Human dose
1g q24h = 67
Change in Log10CFU/g Thigh
Free Ertapenem AUC:MIC*1/tau

CONCLUSIONS

- AUC:MIC per length of the dosing interval (tau), AUC:MIC*1/tau, can be used as an alternative descriptor of time dependency. The value of this target will change depending on the schedule of administration, i.e. if the length of the dosing interval increases from q8h to q24h, then the value of the target will increase 3-fold
- Both fT>MIC and fAUC:MIC*1/tau can be used as pharmacodynamic indices to describe the PD of ertapenem
- A human dose of 1g/day results in drug exposure that produces stasis in this murine model
- Ertapenem is a useful agent to benchmark the development of new antibiotics for antimicrobial resistance

References: [1]Goutelle S, Valour F, Gagnieu MC, Laurent F, Chidiac C, Ferry T. 2018. Population pharmacokinetics and probability of target attainment of ertapenem administered by subcutaneous or intravenous route in patients with bone and joint infection. *J Antimicrob Chemother* **73**:987–994