A Rubio
June 10, 2018
SUN-538

In Vitro Characterization of a Novel Gyrase Inhibitor (SPR719) Against Nontuberculous Mycobacteria

BA Brown-Elliott[1], M Ashcraft[1], G Bush[1], A McKinney[1], K Ritter[1], K Shipp[1], J Mooney[1], RJ Wallace Jr.[1], A Rubio[2]

The University of Texas Health Science Center, Tyler, TX; [2]Spero Therapeutics, Cambridge, MA

Aileen Rubio
Spero Therapeutics
675 Massachusetts Ave
Cambridge, MA 02139
arubio@sperotx.com

ABSTRACT

Background: Nontuberculous mycobacteria (NTM) infections are increasing dramatically globally. *Mycobacterium avium* complex (MAC) and *M. abscessus* are the most frequently encountered NTM among clinical laboratories, and treatment options are extremely limited. In this study, the *in vitro* potency of a novel gyrase inhibitor, SPR719, the microbiologically active form of the orally available prodrug SPR720, was tested against several species of NTM.

Methods: MICs were performed in cation adjusted Mueller Hinton Broth as described and recommended by the Clinical and Laboratory Standards Institute (CLSI M24-A2). Comparator antimicrobials included amikacin, cefoxitin, ciprofloxacin, clarithromycin, doxycycline, imipenem, linezolid, minocycline, moxifloxacin, tigecycline, and trimethoprim/sulfamethoxazole (TMP/SMX) for the rapidly growing species (RGM); amikacin and clarithromycin for the MAC; amikacin, ciprofloxacin, clarithromycin, doxycycline, linezolid, moxifloxacin, rifabutin, rifampin and TMP/SMX for the other slowly growing NTM.

Results: Potency of SPR719 against 154 NTM organisms are found in the table.

Conclusions: SPR719 was found to be potent against multiple clinical strains of NTM with an MIC_{50} range of 0.25 – 4 µg/mL for several species of NTM. These findings support the further advancement of SPR720 for the treatment of NTM disease.

INTRODUCTION

- Infections caused by nontuberculous mycobacteria (NTM) are increasing in prevalence due to improved recognition and diagnosis.
- NTM infections are generally difficult to treat since these organisms are resistant to most of the antituberculous drugs and therefore new agents are needed
- *M. avium* complex (MAC) and the *M. abscessus* complex are the most common pathogens found
- Here we describe the *in vitro* activity of SPR719, a novel gyrase inhibitor, against MAC and *M. abscessus* in comparison to other antibacterial agents

METHODS

- MIC testing of SPR719 was performed by microbroth dilution using Mueller Hinton broth, consistent with M24-A2 CLSI methodology
- Comparator antibiotics included amikacin (AMK), cefoxitin (FOX), ciprofloxacin (CIP), clarithromycin (CLR), doxycycline (DOX), imipenem (IPM), linezolid (LZD), minocycline (MIN), moxifloxacin (MXF), tigecycline (TGC), and trimethoprim/sulfamethoxazole (SXT) for the rapidly growing species (RGM)
- Comparator antibiotics included AMK and CLR for the MAC
- Comparator antibiotics included AMK, CIP, CLR, DOX, LZD, MXF, rifabutin (RFB), rifampin (RIF) and SXT for the other slowly growing NTM

RESULTS

Table 1. SPR719 MIC (µg/mL) values against 161 isolates of nontuberculous mycobacteria

		M. abscessus subsp. *abscessus*	*M. abscessus* subsp. *massiliense*	*M. abscessus / massiliense hybrid*	*M. chelonae*	*M. immunogenum*	*M. fortuitum* group	*M. mucogenicum* group	*M. kansasii*	*M. marinum*	*M. simiae*	*M. avium*	*M. intracellulare*	MAC-X
	N of isolates	33	10	10	10	10	10	10	8	9	10	12	19	10
SPR719	MIC Range	0.25 – 8	0.12 – 4	0.06 – 2	2 – 4	4 – 8	0.06 – 1	0.015 – 0.25	0.002 – 0.015	0.12 – 1	0.5 – 4	0.5 – 2	0.12 – 2	0.12 – 1
	MIC_{50}	2	2	2	4	4	0.25	0.06	0.015	0.5	1	0.5	0.5	1
	MIC_{90}	4	2	2	4	8	1	0.25	0.03	0.5	2	2	2	1
AMK	MIC Range	2 - 64	4 – 16	8 – 16	16 – 32	8 – 16	≤1	≤1	2 - 8	≤1 - 2	8 - 32	8 - >64	8 - >64	8 - 32
	MIC_{50}	8	16	16	16	8	≤1	≤1	4	≤1	16	16	16	8
	MIC_{90}	16	16	16	32	16	≤1	≤1	8	2	16	64	>64	32
FOX	MIC Range	16 - 64	32 - 64	32 – 64	>128	>128	32 - 64	≤4 – 16						
	MIC_{50}	64	64	32	>128	>128	64	8						
	MIC_{90}	64	64	64	>128	>128	64	16						
SXT	MIC Range	4/76 - 8/152	4/76 - 8/152	4/76 - 8/152	≤2/38 - >4/76	2/38 - 8/152	≤0.25/4.75 - 1/19	≤0.25/4.75 -0.5/9.5	≤0.12 - 0.25/4.75	≤0.25/4.75- 1/19	1/19 - 4/76			
	MIC_{50}	4/76	4/76	4/76	≥4/76	4/76	0.5/9.5	≤0.25/4.75	0.25/4.75	0.5/9.5	2/38			
	MIC_{90}	8/152	8/152	8/152	≥4/76	8/152	1/19	0.5/9.5	0.25/4.75	1/19	4/76			
LZD	MIC Range	2 – 32	2 – 16	2 - 16	2 - 16	4 - 32	≤1 – 8	≤1 - 4	≤1 – 4	≤1 - 2	16 - >64	2 - 64	8 - >64	8 - 64
	MIC_{50}	8	8	8	8	16	2	≤1	≤1	2	32	32	32	32
	MIC_{90}	16	16	16	16	32	4	4	4	2	64	64	64	64
CIP	MIC Range	2 - >4	≥4	4 - >4	2 - >4	2 - 8	≤0.12	0.25 – 4	2 - 4	1 - 16	4 - >16			
	MIC_{50}	>4	4	4	≥4	4	≤0.12	0.5	4	8	8			
	MIC_{90}	>4	>4	>4	≥4	8	≤0.12	2	4	16	>16			
IPM	MIC Range	8 – 64	4 - 32	8 - 32	16 - 64	16 - 64	4 - 8	≤2						
	MIC_{50}	16	8	16	16	32	4	≤2						
	MIC_{90}	32	16	32	32	64	8	≤2						
MXF	MIC Range	2 - >8	2 – >8	4 - >8	4 - >8	4 - 8	≤0.25	≤0.25 - 1	≤0.12 - 0.5	0.25 - 4	1 - 8	0.5 - >8	1 - >8	0.5 - >8
	MIC_{50}	8	8	4	4	4	≤0.25	≤0.25	≤0.12	1	4	1	4	4
	MIC_{90}	>8	>8	>8	8	8	≤0.25	0.5	0.5	4	8	8	8	>8
DOX	MIC Range	>8	2 - >16	>16	0.12 - >16	8 - >16	0.12 - >16	≤0.12 - >16	1 - 16	2 – 16	≥16			
	MIC_{50}	>8	>16	>16	>16	>16	0.25	≤.0.12	8	2	≥16			
	MIC_{90}	>8	>16	>16	>16	>16	>16	>16	16	16	>16			
MIN	MIC Range	>16	4 - >8	>8	≤1 - >8	>8	≤1 - >8	≤1 - >8						
	MIC_{50}	>16	8	>8	>8	>8	≤1	≤1						
	MIC_{90}	>16	>8	>8	>8	>8	>8	>8						
TGC	MIC Range	0.06 - 0.5	0.06 - 0.5	0.06 - 0.25	0.06 - 0.5	0.06 - 0.25	0.03 - 0.12	≤0.25 - 0.25						
	MIC_{50}	0.25	0.12	0.25	0.25	0.12	0.06	0.12						
	MIC_{90}	0.5	0.5	0.5	0.5	0.25	0.12	0.25						
CLR	MIC Range	≤2 - >16	≤2	0.12 - 0.25	≤2	≤2	≤0.06 - >16	≤2	0.12 - 0.5	0.5 – 2	8 - >64	0.25 - >64	1 - >64	0.5 - 8
	MIC_{50}	≥16	≤2	0.06	≤2	≤2	16	≤2	0.25	1	8	2	2	2
	MIC_{90}	≥16	≤2	0.5	≤2	≤2	>16	≤2	0.5	2	32	8	>64	4
TOB	MIC Range				≤1 - 4	8 - 16								
	MIC_{50}				≤2	8								
	MIC_{90}				≤2	16								
RFB	MIC Range								≤0.25	≤0.25	4 - >8			
	MIC_{50}								≤0.25	≤0.25	8			
	MIC_{90}								≤0.25	≤0.25	>8			
RIF	MIC Range								≤0.12-1	0.5-1	>8			
	MIC_{50}								0.25	0.5	>8			
	MIC_{90}								1	1	>8			

- Comparator MICs were within usual ranges for each species tested
- For NTM tested, including isolates R to CIP or MXF, MICs for SPR719 were at least 1 dilution lower than CIP or MXF
- MIC_{50} for all RGM (n=93) tested was <u><</u>4 µg/mL
- MIC_{90} for MAC (n=41), including 10 isolates of MAC-X, was <u><</u>2 µg/mL
- Quality control performed by RGM with *M. peregrinum* ATCC 700686 was 0.25 – 0.5 µg/mL and *Staphylococcus aureus* <0.015 – 0.06 µg/mL
- Quality control performed for slowly growing NTM with *M. marinum* ATCC 927 was 0.12 – 0.5 µg/mL

Figure 1. Structure of SPR719

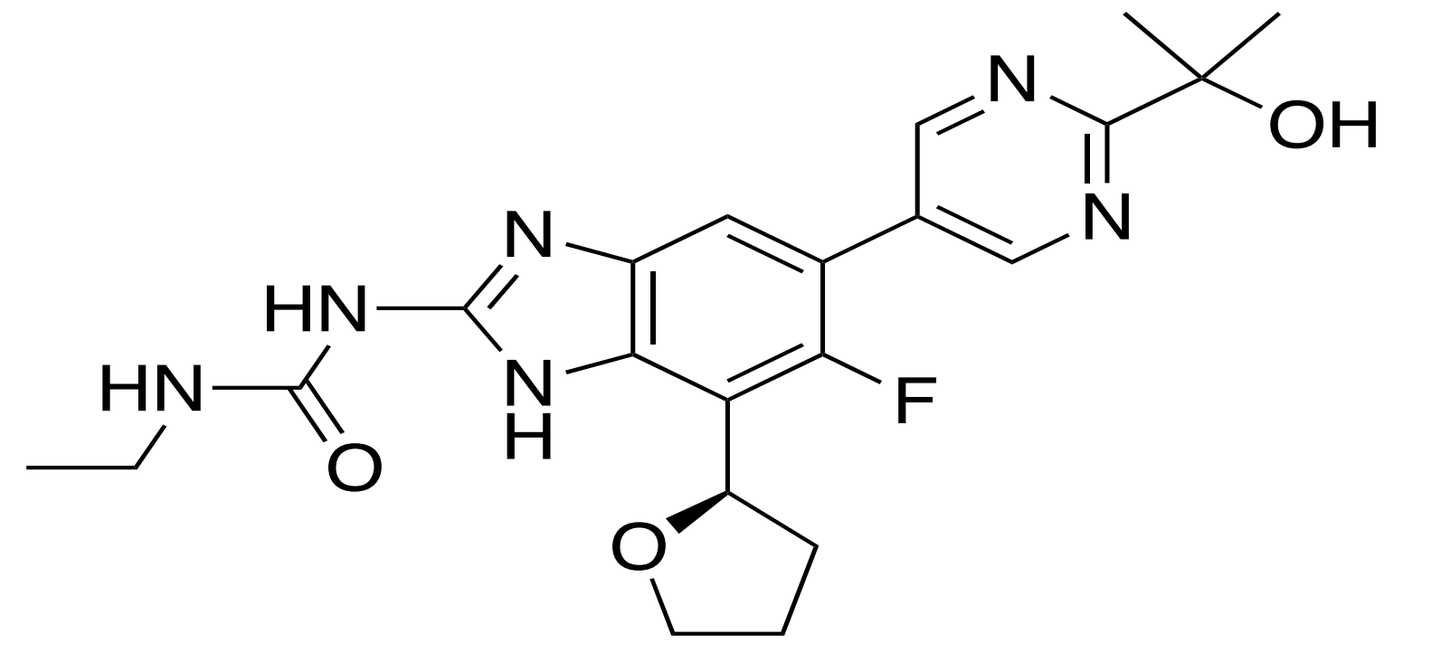
N
OH
N
N
HN
F
HN
N
H
O
O

CONCLUSIONS

- SPR719 was found to be potent against most clinical strains of NTM with an MIC_{50} range of 0.25–4 µg/mL for several clinically significant species of NTM
- SPR719 may provide a useful treatment alternative to quinolones especially among species that are intrinsically R to quinolones (e.g., *M. abscessus* complex, *M. immunogenum, M. simiae,* and most isolates of the MAC)
- These findings support the further advancement of SPR720 for the treatment of NTM disease

ACKNOWLEDGEMENTS

The authors wish to thank Spero Therapeutics for funding this study. We also thank the mycobacterial molecular identification team at the University of Texas Health Science Center at Tyler, including Ravikiran Vasireddy, Sruthi Vasireddy, Adrian Almodovar, Elena Iakhiaeva, and Terry Smith for sequencing the isolates and Joanne Woodring for her clerical support.

REFERENCE

CLSI, M24-A2, 2011.

UTHealth
The University of Texas Health Science Center at Tyler