Comparison of Healthcare Resource Utilization Among Adult Patients Treated with Omadacycline for ABSSSI or CABP in the 30 Day Pre- and Post-Omadacycline Prescription

Thomas P. Lodise, PharmD, PhD[1]; Kyle Gunter, PharmD, MBA, BCPPS[2]; Mauricio Rodriguez, PharmD, BCPS, BCCCP, BCIDP[2]; Fan Mu, PhD[3]; Holly Hoffman-Roberts, PharmD[2]; Emily Gao, MPH[3]; Danni Yang, BA[3]; Steve Sandor, JD, MBA[2]; Gail Berman, MD[2]

[1] Albany College of Pharmacy and Health Sciences, Albany, New York; [2] Paratek Pharmaceuticals, Inc., King of Prussia, PA, USA; [3] Analysis Group, Inc., Boston, MA, USA

Background

Community-acquired bacterial pneumonia (CABP) and acute bacterial skin and skin structure infections (ABSSSI) continue to be a substantial cause of morbidity and mortality in the United States[1–3]

Ambulatory visits for CABP and ABSSSI are ~4.5 million and 8.4 million per year in the United States respectively[4–6]

Annual medical costs linked to pneumonia exceed $17 billion, and among the Medicare population, the burden of CABP costs was estimated to be $13 billion in 2008.[7] Limited information is available on the burden of ABSSSI

Omadacycline, a first in class oral and intravenous (IV) once-daily antibiotic, recently approved for the treatment of adult patients with CABP and ABSSSI, with activity against key pathogens, S. pneumoniae and S. aureus including drug resistant strains

Methods

US claims data from the PatientSource® database, supplied by Source Healthcare Analytics, LLC, a Symphony Health Solutions Corporation covering ~274 million patients were used for this study, including data from October 2018 to September 2020

A pre-post study design was selected to assess how patient outcomes changed pre vs. post-omadacycline prescription

Patients treated with omadacycline (index date: prescription date) for ABSSSI or CABP who had 3 months of data available prior to the index date (baseline period) and 1 month of data following the index date (study period) were included in the study

Patients were classified as complicated and uncomplicated based on infection history

Statistical analysis

- Baseline characteristics and treatment characteristics were compared using ANOVA tests for continuous variables and Chi-squared tests for categorical variables
- Healthcare resource use (HRU), including all-cause and disease-specific visits, were assessed and compared between the pre- and post-periods. P-values were generated using generalized estimating equation (GEE) regression with robust sandwich estimators to account for within-patient correlation

Table 1. Baseline characteristics among ABSSSI patients in the 90-day pre-omadacycline prescription period

	Complicated ABSSSI (N = 83)	Non-complicated ABSSSI (N = 106)
Demographics		
Age on index date in years, mean ± SD	58.11 ± 13.46	57.81 ± 15.98
Female, n (%)	37 (44.6)	63 (59.4)
Region, n (%)		
South	37 (44.6)	49 (46.2)
Midwest	21 (25.3)	28 (26.4)
Northeast	15 (18.1)	20 (18.9)
West	10 (12.0)	8 (7.6)
Unknown	0 (0.0)	1 (0.9)
Clinical characteristics		
CCI	1.64 ± 1.68	1.05 ± 1.44
Comorbidities of interest, n (%)		
Osteomyelitis	44 (53.0)	0 (0.0)
Diabetes	43 (51.8)	35 (33.0)
Septicemia/sepsis/bacteremia	27 (32.5)	0 (0.0)
Peripheral vascular disease	27 (32.5)	10 (9.4)
Renal diseases	21 (25.3)	18 (17.0)
Chronic obstructive pulmonary disease	17 (20.5)	20 (18.9)
Implant/graft/prosthetic joint infection	17 (20.5)	0 (0.0)
Congestive heart failure	14 (16.9)	11 (10.4)
Urinary tract infection	10 (12.0)	3 (2.8)
Surgical site infection	7 (8.4)	7 (6.6)
At least one non-Nuzyra antibiotic in the baseline period	68 (81.9)	82 (77.4)

Table 2. Baseline characteristics among CABP patients in the 90-day pre-omadacycline prescription period

	Complicated CABP (N = 20)	Non-complicated CABP (N = 49)
Demographics		
Age on index date in years, mean ±SD	64.30 ± 17.34	63.18 ± 14.61
Female, n (%)	11 (55.0)	30 (61.2)
Region, n (%)		
South	18 (90.0)	38 (77.5)
Midwest	1 (5.0)	4 (8.2)
Northeast	0 (0.0)	4 (8.2)
West	1 (5.0)	3 (6.1)
Unknown	0 (0.0)	0 (0.0)
Clinical characteristics		
CCI	2.30 ± 1.98	1.63 ± 1.74
Comorbidities of interest, n (%)		
Septicemia/sepsis/bacteremia	16 (80.0)	0 (0.0)
Chronic obstructive pulmonary disease	11 (55.0)	24 (49.0)
Renal diseases	8 (40.0)	9 (18.4)
Congestive heart failure	6 (30.0)	8 (16.3)
Complicated pneumonia	5 (25.0)	0 (0.0)
Diabetes	5 (25.0)	6 (12.2)
Bacterial pneumonia (other than CABP)	3 (15.0)	7 (14.3)
Rheumatic diseases	3 (15.0)	3 (6.1)
Peripheral vascular disease	2 (10.0)	5 (10.2)
Urinary tract infection	2 (10.0)	2 (4.1)
Any malignancy	2 (10.0)	3 (6.1)
Mild liver disease	1 (5.0)	3 (6.1)
Clostridioides difficile infection	1 (5.0)	3 (6.1)
At least one non-Nuzyra antibiotic in the baseline period	20 (100.0)	43 (87.8)

Funding and disclosures: TPL: Consultant and Speaker– Paratek Pharmaceuticals, Inc. KG, HH-R, SS, GB: Employee and shareholder – Paratek Pharmaceuticals, Inc. MR was an employee and shareholder of Paratek Pharmaceuticals, Inc. at time of study. FM, EG, DY: Employee – Analysis Group, Inc. This study was funded by Paratek Pharmaceuticals, Inc.

HRU reductions after treatment with omadacycline for both ABSSSI and CABP

Objectives

To address the evidence data gap around real-world use of omadacycline, this study had the following objectives:

- To understand patient and treatment characteristics among those with an omadacycline prescription for CABP and ABSSSI
- To assess healthcare resource use associated with the use of omadacycline for CABP and ABSSSI in the pre- and post-omadacycline prescription period

Conclusions

This study provided the first real world characterization of patients treated with omadacycline for ABSSSI or CABP, including patients with high comorbidity burden and extensive use of antibiotics

After receiving omadacycline, patients had lower all-cause and disease-specific HRU, including inpatient visits, in the 30-day post-omadacycline prescription period relative to the 30-day pre-omadacycline prescription period

HRU reductions after treatment with omadacycline were consistent among complicated and non-complicated patients for both ABSSSI and CABP

Click here for a copy of this poster

Click here for additional information

PARATEK

Results

The study population selection is shown in **Figure 1**

A high proportion of patients in both the ABSSSI and CABP cohorts experienced prior bacterial infections in the baseline period and patients had a high comorbidity burden (**Tables 1 and 2**)

In the baseline period, most patients were treated with at least one prior antibiotic class

All-cause and disease-specific HRU showed significant reduction in the 30-day post-omadacycline prescription vs. 30-day pre-omadacycline prescription (**Figures 2 and 3**)

Figure 1. Sample Selection

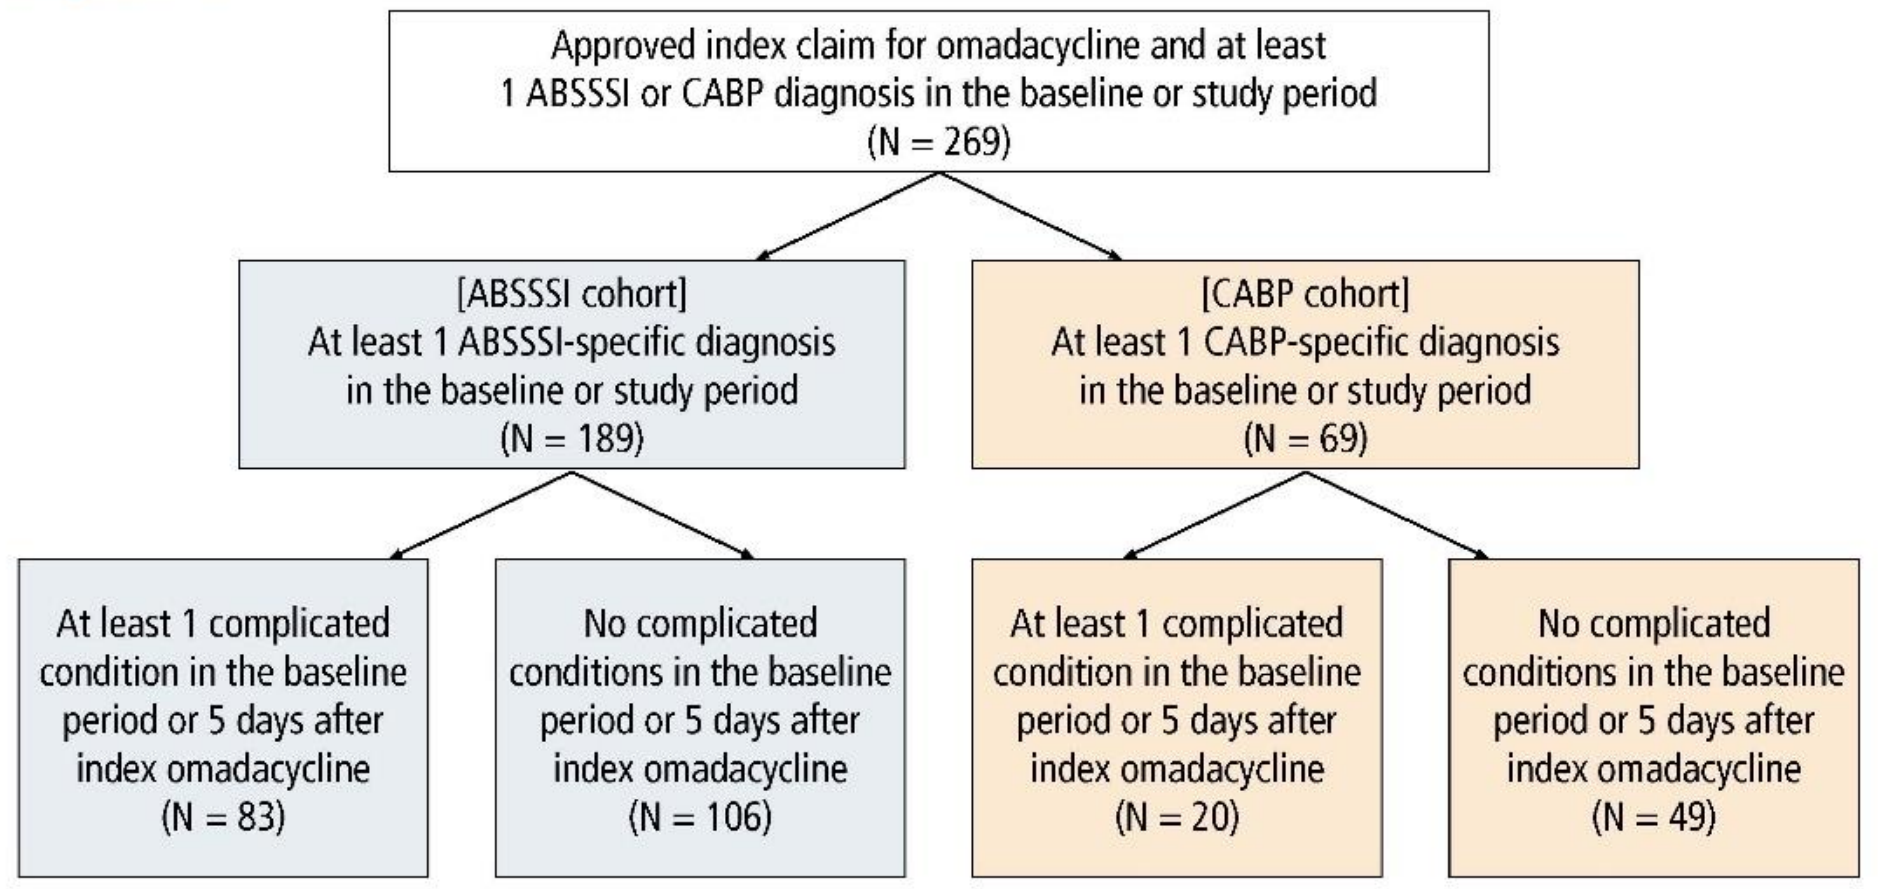
Approved index claim for omadacycline and at least 1 ABSSSI or CABP diagnosis in the baseline or study period (N = 269)
[ABSSSI cohort] At least 1 ABSSSI-specific diagnosis in the baseline or study period (N = 189)
[CABP cohort] At least 1 CABP-specific diagnosis in the baseline or study period (N = 69)
At least 1 complicated condition in the baseline period or 5 days after index omadacycline (N = 83)
No complicated conditions in the baseline period or 5 days after index omadacycline (N = 106)
At least 1 complicated condition in the baseline period or 5 days after index omadacycline (N = 20)
No complicated conditions in the baseline period or 5 days after index omadacycline (N = 49)

(Definition of complicated cohorts: ABSSSI: osteomyelitis, bone/joint infection, sepsis/bacteremia, endocarditis, implant infection, necrotizing fasciitis, meningitis; CABP: severe pneumonia, lung abscess, sepsis/bacteremia, endocarditis, meningitis).

Figures 2a and 2b. Comparison of HRU in the 30-day pre-omadacycline prescription vs. 30-day post-omadacycline prescription period among ABSSSI patients

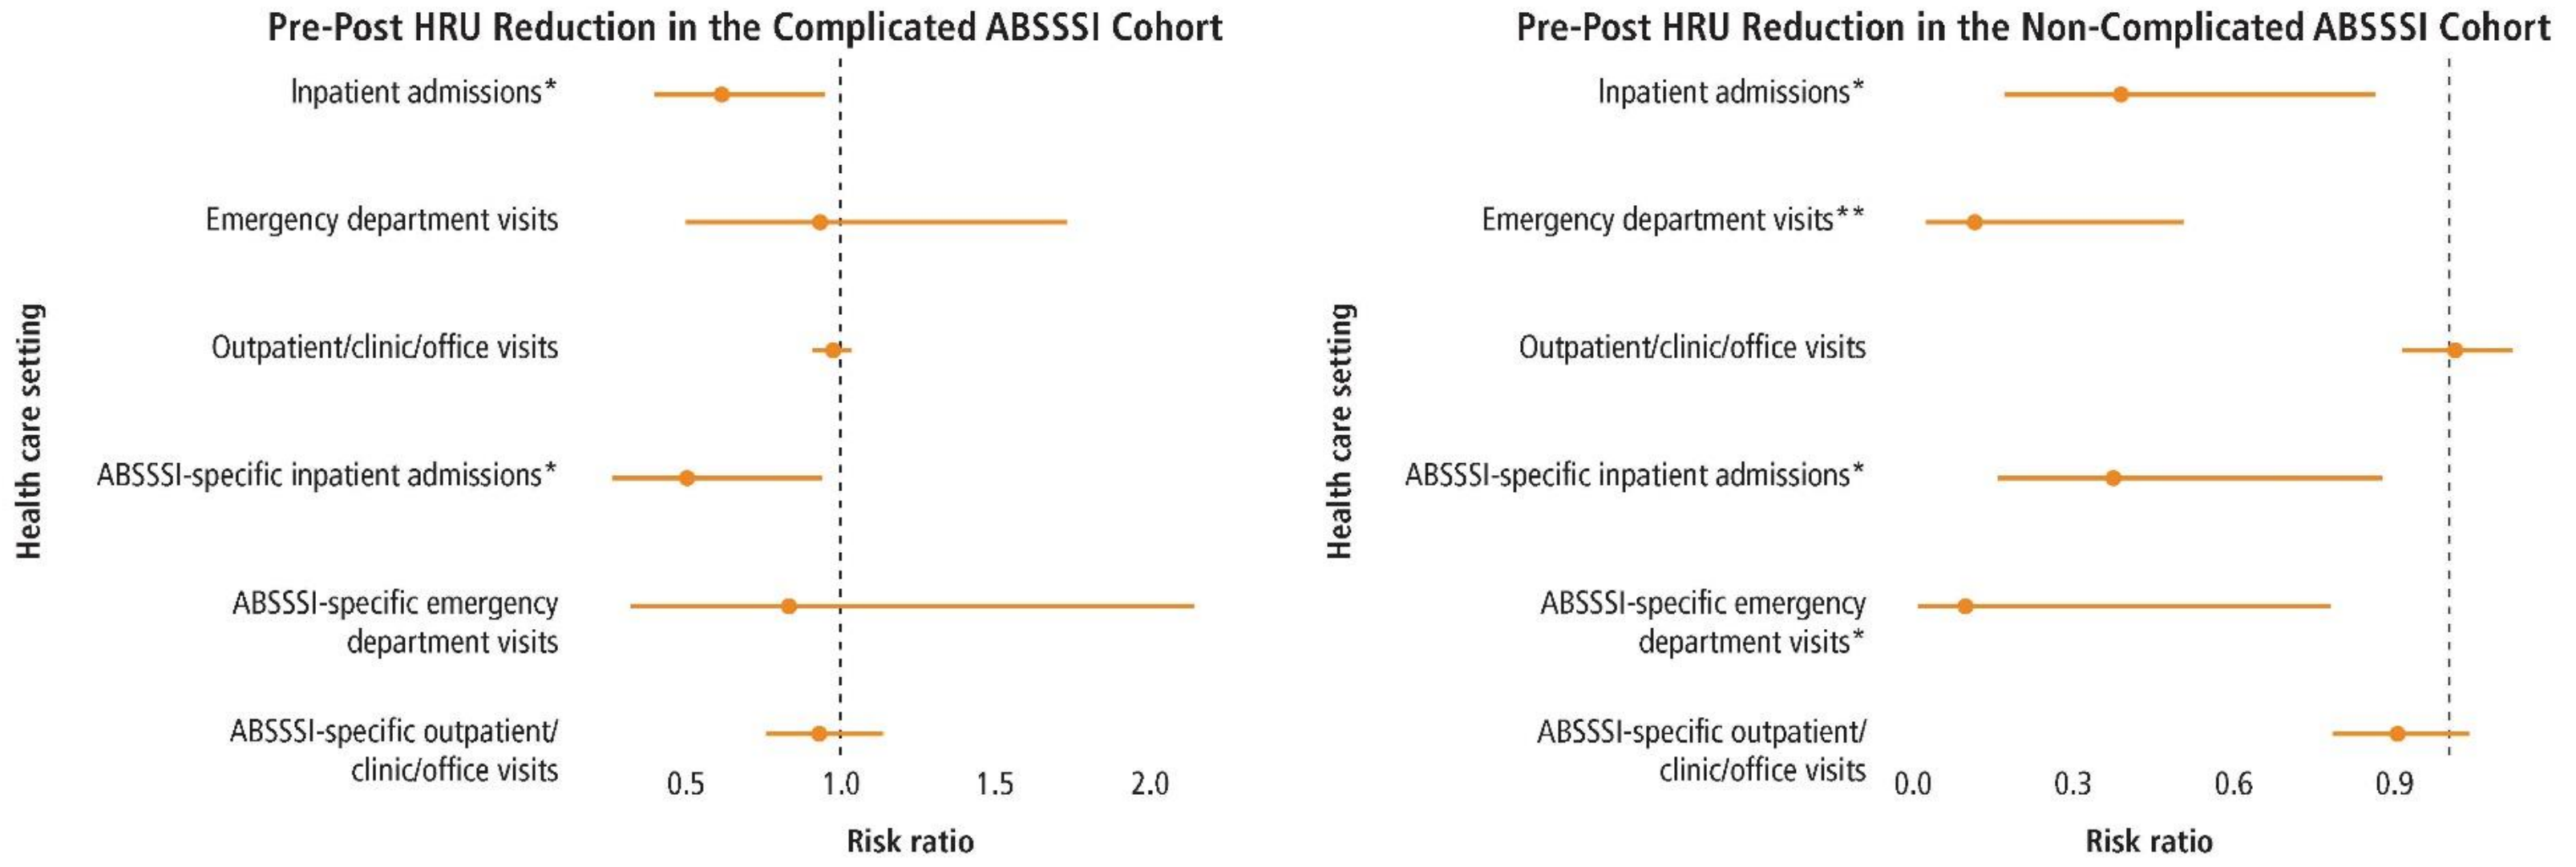
Pre-Post HRU Reduction in the Complicated ABSSSI Cohort
Inpatient admissions*
Emergency department visits
Outpatient/clinic/office visits
ABSSSI-specific inpatient admissions*
ABSSSI-specific emergency department visits
ABSSSI-specific outpatient/ clinic/office visits
Health care setting
0.5
1.0
1.5
2.0
Risk ratio
Pre-Post HRU Reduction in the Non-Complicated ABSSSI Cohort
Inpatient admissions*
Emergency department visits**
Outpatient/clinic/office visits
ABSSSI-specific inpatient admissions*
ABSSSI-specific emergency department visits*
ABSSSI-specific outpatient/ clinic/office visits
Health care setting
0.0
0.3
0.6
0.9
Risk ratio

*p-value < 0.05, **p-value < 0.01, ***p-value < 0.001

Figures 3a and 3b. Comparison of HRU in the 30-day pre-omadacycline prescription vs. 30-day post-omadacycline prescription period among CABP patients

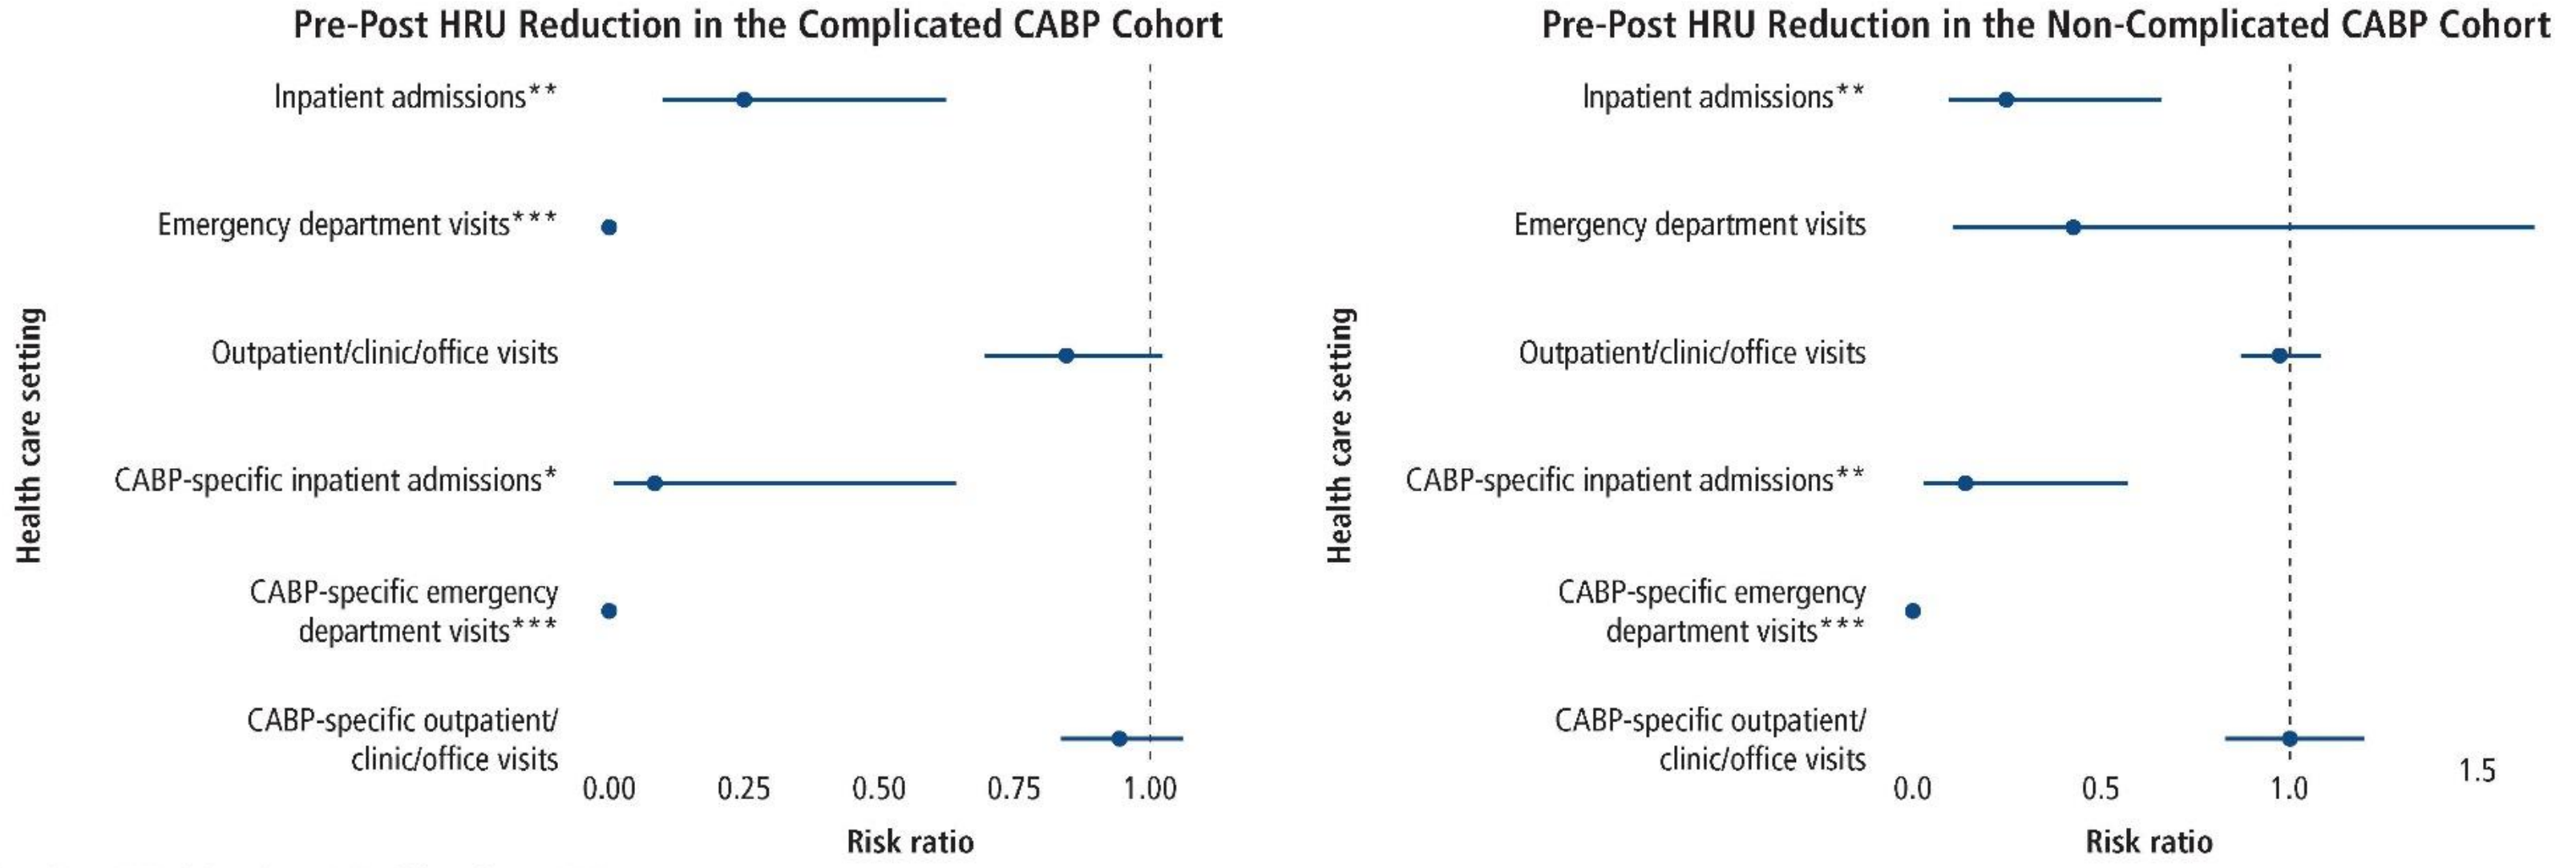
Pre-Post HRU Reduction in the Complicated CABP Cohort
Inpatient admissions**
Emergency department visits***
Outpatient/clinic/office visits
CABP-specific inpatient admissions*
CABP-specific emergency department visits***
CABP-specific outpatient/ clinic/office visits
Health care setting
0.00
0.25
0.50
0.75
1.00
Risk ratio
Pre-Post HRU Reduction in the Non-Complicated CABP Cohort
Inpatient admissions**
Emergency department visits
Outpatient/clinic/office visits
CABP-specific inpatient admissions**
CABP-specific emergency department visits***
CABP-specific outpatient/ clinic/office visits
Health care setting
0.0
0.5
1.0
1.5
Risk ratio

*p-value < 0.05, **p-value < 0.01, ***p-value < 0.001

Presented at the 5th Annual Texas Medical Center Antimicrobial Resistance & Stewardship Conference [Virtual], January 19–21, 2022