Potential Cost-Saving Opportunities With Early Use of Omadacycline in Hospitalized Patients With ABSSSI With Inadequate Early Response to Vancomycin Plus Piperacillin-Tazobactam: Findings From a Decision-Analytic Budget Impact Model

Kenneth LaPensee, PhD, MPH[1]; Thomas Lodise, PharmD, PhD[2]; Camelia Graham, PhD, MSPH[3]; Hoa Van Le, MD, PhD[3]; Canter Martin, MBA[3]; Rohit Mistry, MSc[3]; Kate Young, PhD, MPH[3]

[1]Paratek Pharmaceuticals, Inc., King of Prussia, PA, USA; [2]Albany College of Pharmacy and Health Sciences, Albany, NY, USA; [3]PAREXEL Access Consulting, London, UK.

BACKGROUND

- Care of patients with acute bacterial skin and skin structure infections (ABSSSI) places a major financial burden on the US healthcare system[1]
- Average inpatient costs per ABSSSI patient range between $6,000 to $13,000 2010 US dollars (USD), with multi-day room and board expenses comprising 50% of total costs[2]
- Vancomycin (VAN) with piperacillin-tazobactam (TZP) is a commonly prescribed antibiotic regimen with hospitalized patients with ABSSSI[3]
- However, recent real-world evidence analysis suggests up to 29.5% of patients with ABSSSI receiving VAN monotherapy and 46.1% using a combination of VAN plus TZP have an inadequate early response[3]
 - Patients with an inadequate early response had substantially increased LOS (5.4-day increase), antibiotic treatment duration (increase of 3.7 days), and cost of care (1.4-fold increase) for patients with ABSSSI
- OMC, a once-daily antibiotic with broad spectrum activity, including MRSA, is currently under development for IV and oral treatment of patients with ABSSSI
- Antibiotics with intravenous (IV) and oral (PO) dosing formulations have been shown to shorten hospital stay across several infection types, including ABSSSI[4]
 - A pan-European study found that patients with MRSA-cSSTI who switched from IV to oral antibiotic therapy had 5.3 fewer IV days ($P<0.001$) and 1.9 fewer inpatient days ($P=0.162$) compared with those who received IV therapy for the entire treatment duration[5]

OBJECTIVES

- To explore the management of hospitalized patients with ABSSSI who have an inadequate early response to VAN-based regimens. We compared maintaining these patients on VAN plus TZP vs early switch at Day 4 to OMC
- To determine the economic impact of 1 and 2-day reductions in hospital LOS with OMC relative to continuing on VAN plus TZP

METHODS

- A budget impact model was developed in Microsoft Excel 2010® based on current clinical practice to simulate treatment of patients hospitalized with ABSSSI who receive VAN plus TZP
 - The analysis estimates the budget impact of using OMC per 100 simulated ABSSSI patients who have inadequate early response to VAN plus TZP from a hypothetical US hospital perspective
 - Since the incidence of ABSSSI varies by hospital, a population size of 100 patients has been utilized to facilitate comparison with individual hospitals
 - This analysis assumes patients with ABSSSIs who fail to achieve cessation of lesion spread and absence of fever after 72 hours of initiation of antibiotic therapy are inadequate early responders[3]
- As illustrated in **Figure 1**, patients hospitalized with ABSSSI enter the model following 3 days of inpatient treatment with IV VAN plus TZP
- After this initial treatment, these patients are defined as inadequate early responders and may be treated in one of two routes: continue treatment with IV VAN plus TZP until response (as described in Amara 2013)[3] or inpatient treatment switch to OMC (IV) at Day 4
- Three scenarios for OMC treatment were analyzed to further investigate the impact of this assumption:
 - Patients receive OMC IV treatment in the hospital for 5 days (base case)
 - Patients are switched to an oral discharge treatment regimen and discharged 1 or 2 days earlier

Figure 1. Patient flow in the budget impact model.

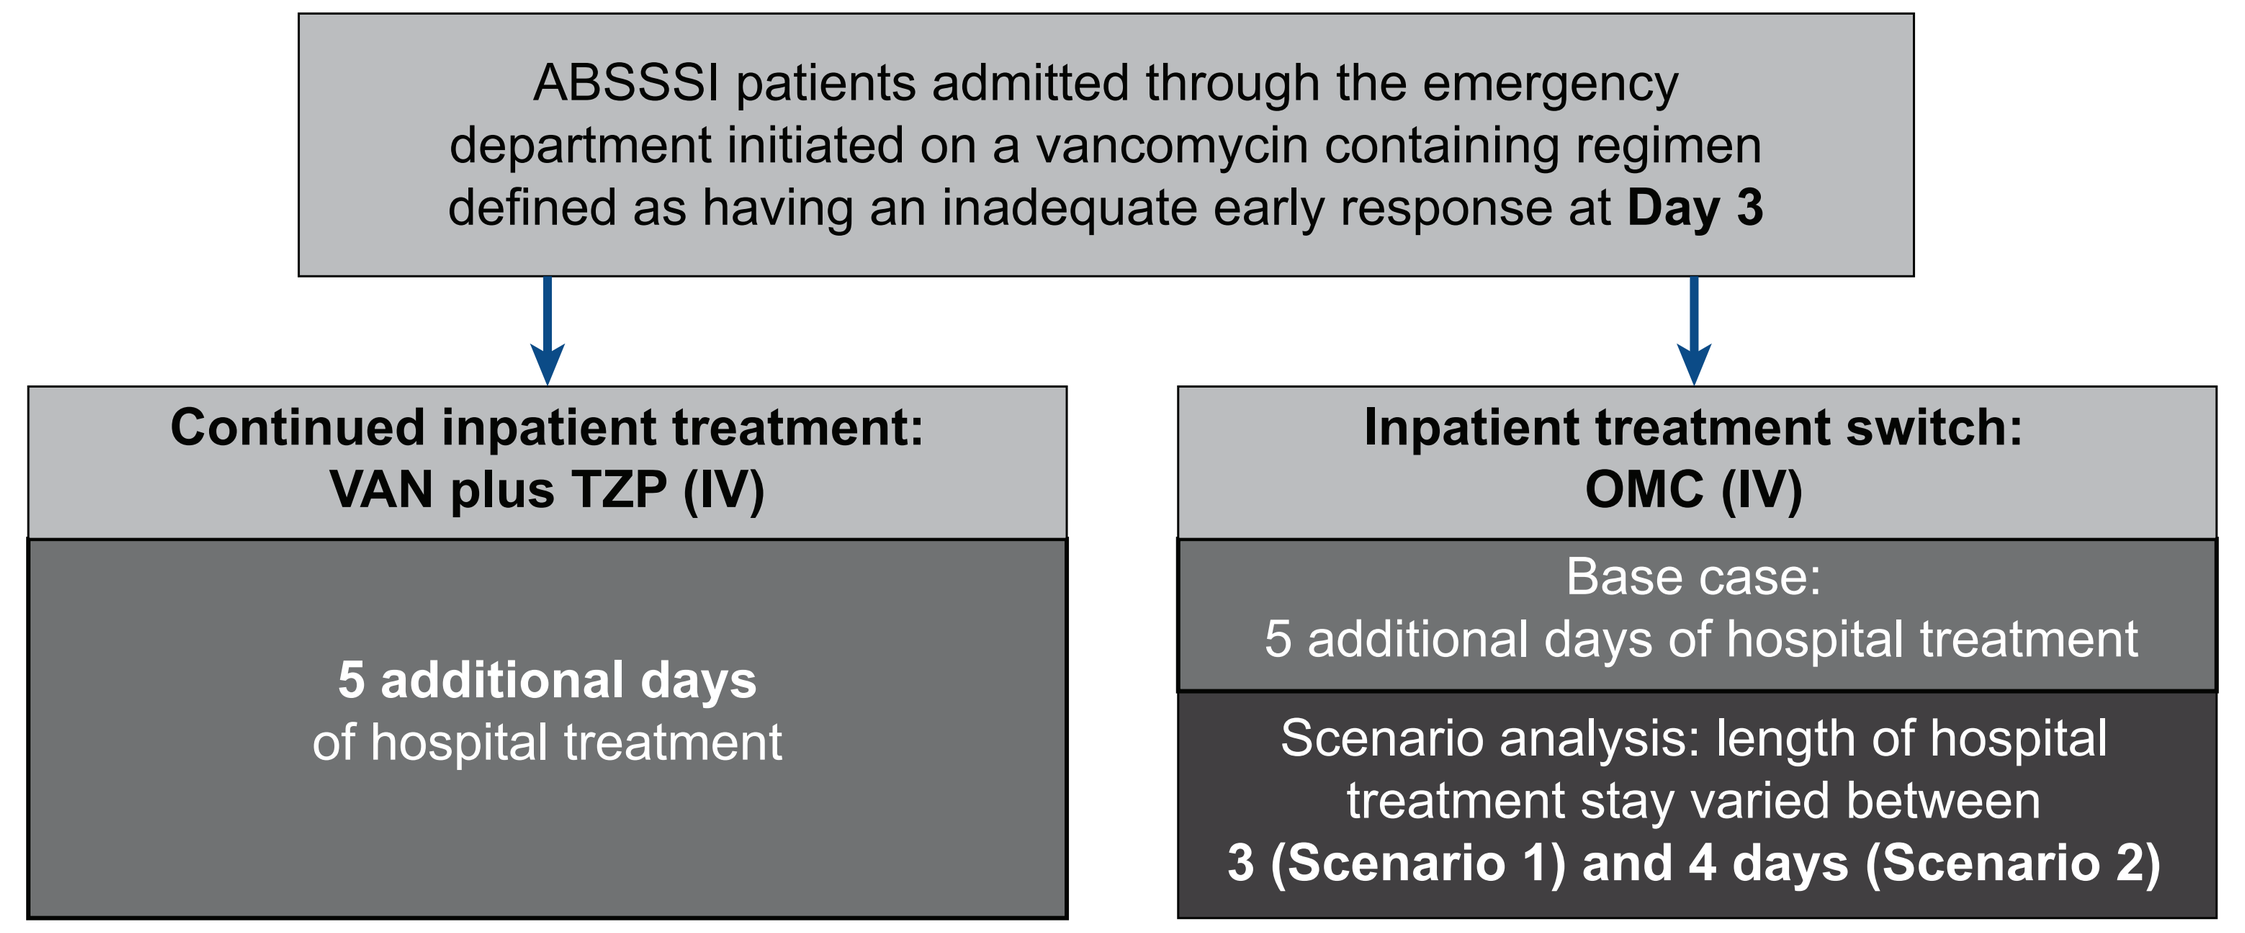
ABSSSI patients admitted through the emergency department initiated on a vancomycin containing regimen defined as having an inadequate early response at Day 3
Continued inpatient treatment: VAN plus TZP (IV)
5 additional days of hospital treatment
Inpatient treatment switch: OMC (IV)
Base case: 5 additional days of hospital treatment
Scenario analysis: length of hospital treatment stay varied between 3 (Scenario 1) and 4 days (Scenario 2)

Definition of inadequate early response

For the purpose of evaluating drug treatment, early clinical response (defined as cessation of lesion spread and absence of fever) is assessed at 48 to 72 hours after initiating antibiotic therapy (FDA, 2010). As such, 72-hour response rates have been assessed in clinical trials evaluating antimicrobial therapy in ABSSSI patients[6]

METHODS

Modeling Assumptions

- Patients enter the analysis following 3 days of treatment with IV VAN plus TZP when they meet the criteria for inadequate clinical early response[3]
- The patient population is assumed to have an ABSSSI without identifiable pathogens
- The analysis excludes occurrence and treatment of AEs
- A 100% cure rate is assumed at the end of treatment; no therapies after the switch to OMC or continued VAN plus TZP are considered
- The population consists of a 100% Medicare insured population (2016 USD)
- Outpatient costs have been excluded from the analysis in line with cost consideration of US hospital payers

Hospital Cost Inputs

- The analysis considers cost of drug acquisition and selected drug monitoring (**Table 1**)
- Cost context considers direct medical costs, including costs reimbursed under Medicare
- As a conservative estimate, administration and standard monitoring costs are assumed to be included in the inpatient cost per day (room and board plus services)
 - The only exception is VAN assays, as this was deemed a significant treatment-specific monitoring requirement
- Wholesale acquisition costs (WAC) are used for treatment acquisition (Medi-Span® Price Rx®),[7] with the exception of OMC, which are simulated, since OMC has not been commercialized. The stated acquisition cost for OMC does not include any potential manufacturer discounts
 - The price of OMC is varied during a sensitivity analysis between $300 and $600 per day
- All costs are inflated to 2016 values using the medical consumer price index as reported by the Bureau of Labor Statistics[8]

Table 1. Cost Inputs

Daily Treatment Acquisition Costs for ABSSSI					
Comparator	**Dose**	**Formulation**	**Vial Size**	**WAC per Vial, Pack, or Bottle**	**Source**
OMC	100 mg every 24 h	IV: 100 mg	100 mg	$300, $450, $600	N/A
VAN	1 g every 12 h	IV: 10 g reconstituted	10 g	$33.00	Medi-Span® Price Rx®[7]
TZP	3.375 g every 6 h	IV: 4/0.5 g reconstituted	10 vials of 4/0.5 g	$76.00	
Cost of Care					
Item				**Medicare Price**	**Source**
Inpatient cost, per day (room & board + services)				$1,358.84	Premier Hospital Database[9] (2010 inflated to 2016 costs)
VAN trough assay				$18.46 per assay	Medicare, Laboratory Fee Schedule, 2016[10]

Abbreviations: ABSSSI, acute bacterial skin and skin structure infection; g, grams; h, hour; IV, intravenous; mg, milligrams; OMC, omadacycline; PO, per os (by mouth); TZP, piperacillin-tazobactam; VAN, vancomycin; WAC, wholesale acquisition cost.

RESULTS

- Early discharge by 1 or 2 days with OMC can be cost-saving; the degree varies by the simulated per diem drug acquisition costs (**Table 2**, **Figure 2**)
- OMC is cost-saving at $300/day when LOS is shortened by 1 day
- All OMC costs used in the analysis are cost-saving when LOS is shortened by 2 days
- Depending on whether patients are discharged 1 or 2 days earlier, higher acquisition costs may still demonstrate budget impact saving

Table 2. Scenario Analysis Incremental Results per 100 Patients Admitted (Scenario Assessed: Additional Length of Stay Following Inadequate Early Responder Status)

Scenario	OMC	VAN Plus TZP	Reduction in LOS vs VAN Plus TZP
Base case	5 days	5 days	0 days
Scenario 1	4 days	5 days	–1 day
Scenario 2	3 days	5 days	–2 days

Abbreviations: OMC, omadacycline; TZP, piperacillin-tazobactam; VAN, vancomycin.

Figure 2. Incremental budget impact with reduction in LOS achieved with switch to OMC compared with VAN plus TZP.

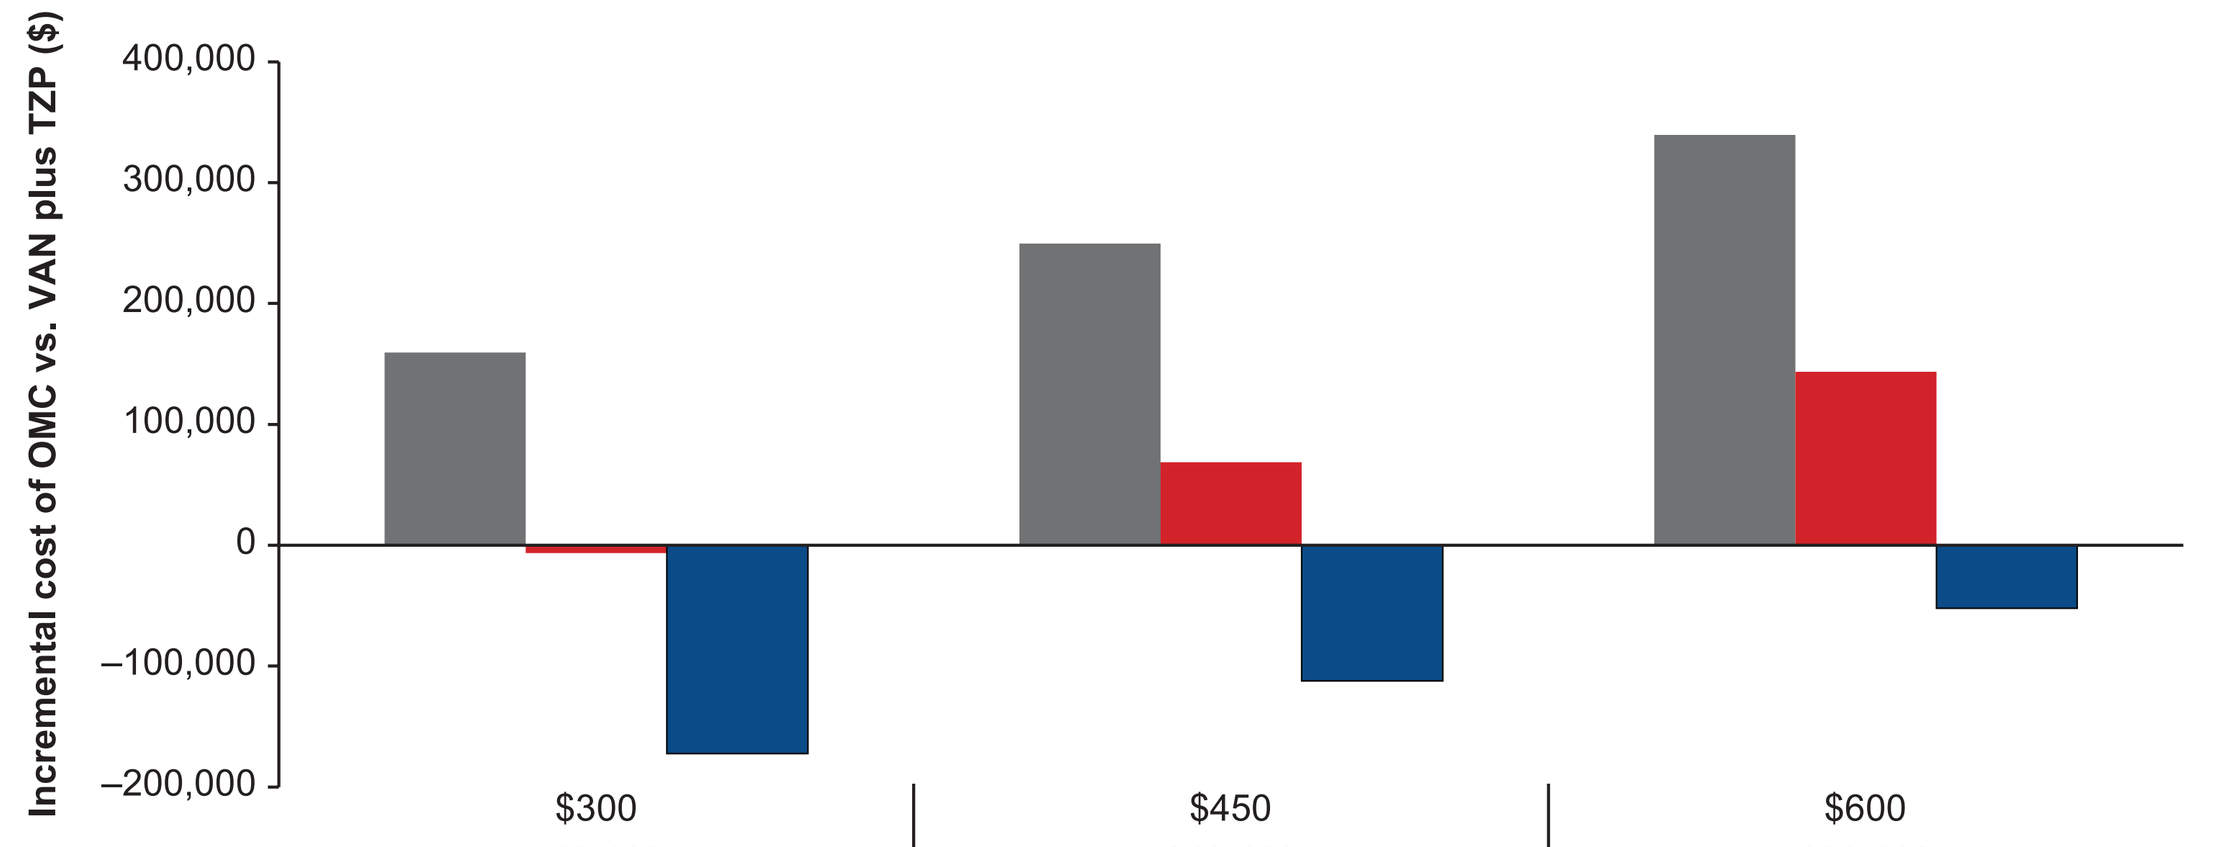
Incremental cost of OMC vs. VAN plus TZP ($)
400,000
300,000
200,000
100,000
0
–100,000
–200,000

	$300	$450	$600
Base case	159,468	249,468	339,468
Scenario 1	–6,416	68,584	143,584
Scenario 2	–172,299	–112,299	–52,299

OMC acquisition cost per day ($)

RESULTS

- The per diem cost-savings to a hospital for treating 100 patients with ABSSSI who have an inadequate early response to VAN plus TZP are the largest contributor to the cost-savings and offset the increased drug acquisition cost of OMC (**Figures 3-5**)

Figure 3. Disaggregated costs for treatment after inadequate response with 5 days of OMC or 5 days of VAN plus TZP.

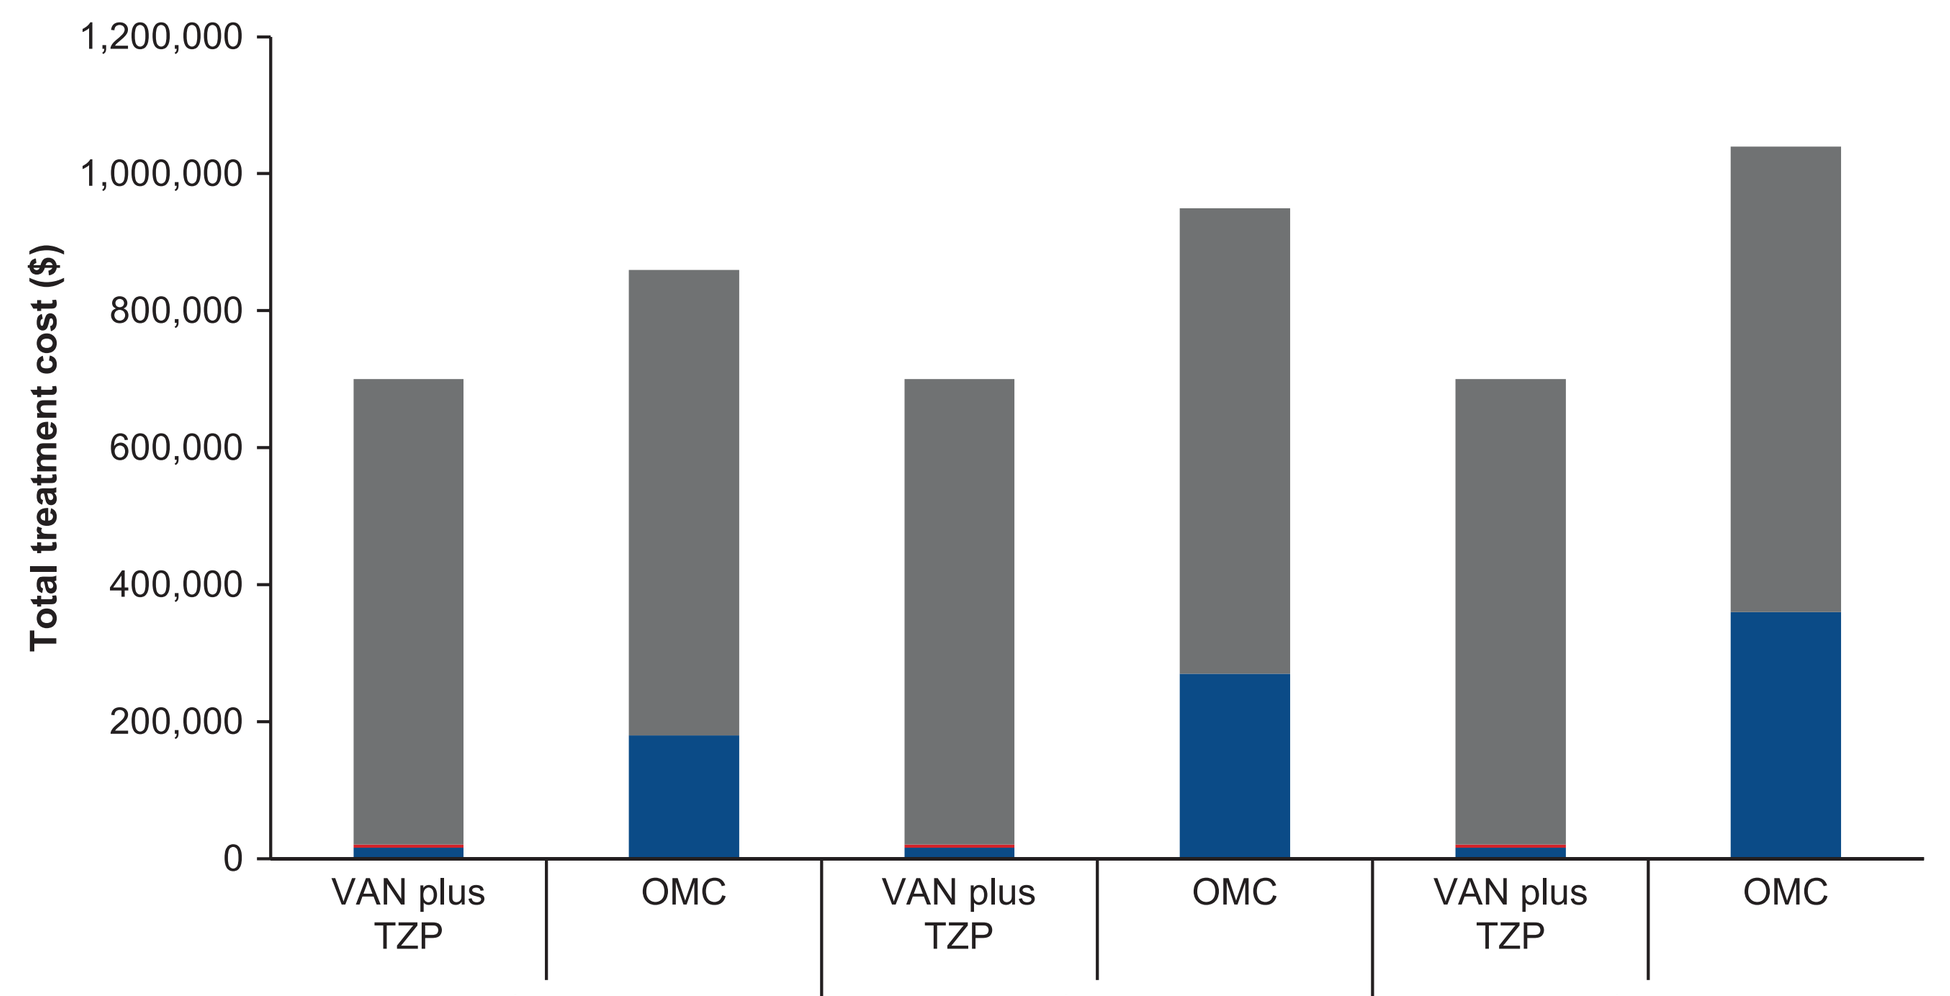
Total treatment cost ($)
1,200,000
1,000,000
800,000
600,000
400,000
200,000
0

	$300 VAN plus TZP	$300 OMC	$450 VAN plus TZP	$450 OMC	$600 VAN plus TZP	$600 OMC
Inpatient costs	679,419	679,419	679,419	679,419	679,419	679,419
Monitoring cost	4,362	0	4,362	0	4,362	0
Drug acquisition cost	16,170	180,000	16,170	270,000	16,170	360,000

OMC acquisition cost per day ($)

Abbreviations: OMC, omadacycline; TZP, piperacillin-tazobactam; VAN, vancomycin.

Figure 4. Disaggregated costs for treatment after inadequate response with 4 days of OMC or 5 days of VAN plus TZP (1 day LOS reduction).

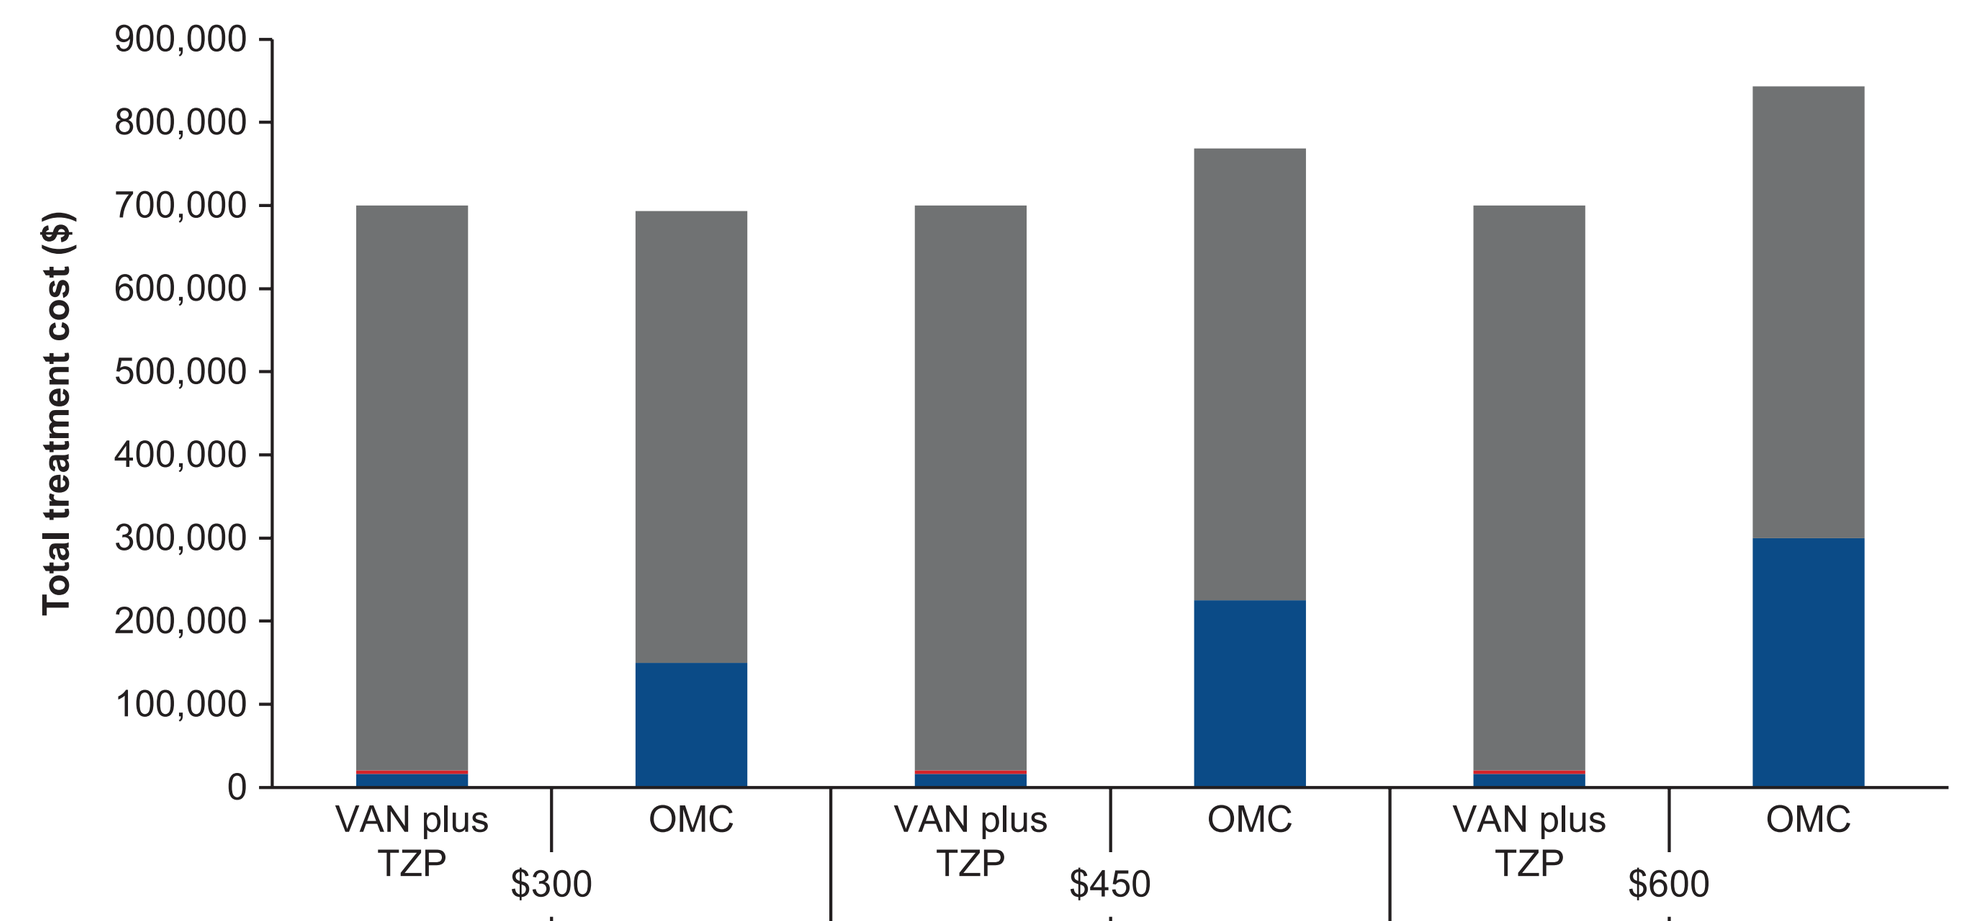
Total treatment cost ($)
900,000
800,000
700,000
600,000
500,000
400,000
300,000
200,000
100,000
0

	$300 VAN plus TZP	$300 OMC	$450 VAN plus TZP	$450 OMC	$600 VAN plus TZP	$600 OMC
Inpatient costs	679,419	543,535	679,419	543,535	679,419	543,535
Monitoring cost	4,362	0	4,362	0	4,362	0
Drug acquisition cost	16,170	150,000	16,170	225,000	16,170	300,000

OMC acquisition cost per day ($)

Abbreviations: LOS, length of stay; OMC, omadacycline; TZP, piperacillin-tazobactam; VAN, vancomycin.

Figure 5. Disaggregated costs for treatment after inadequate response with 3 days of OMC or 5 days of VAN plus TZP (2 day LOS reduction).

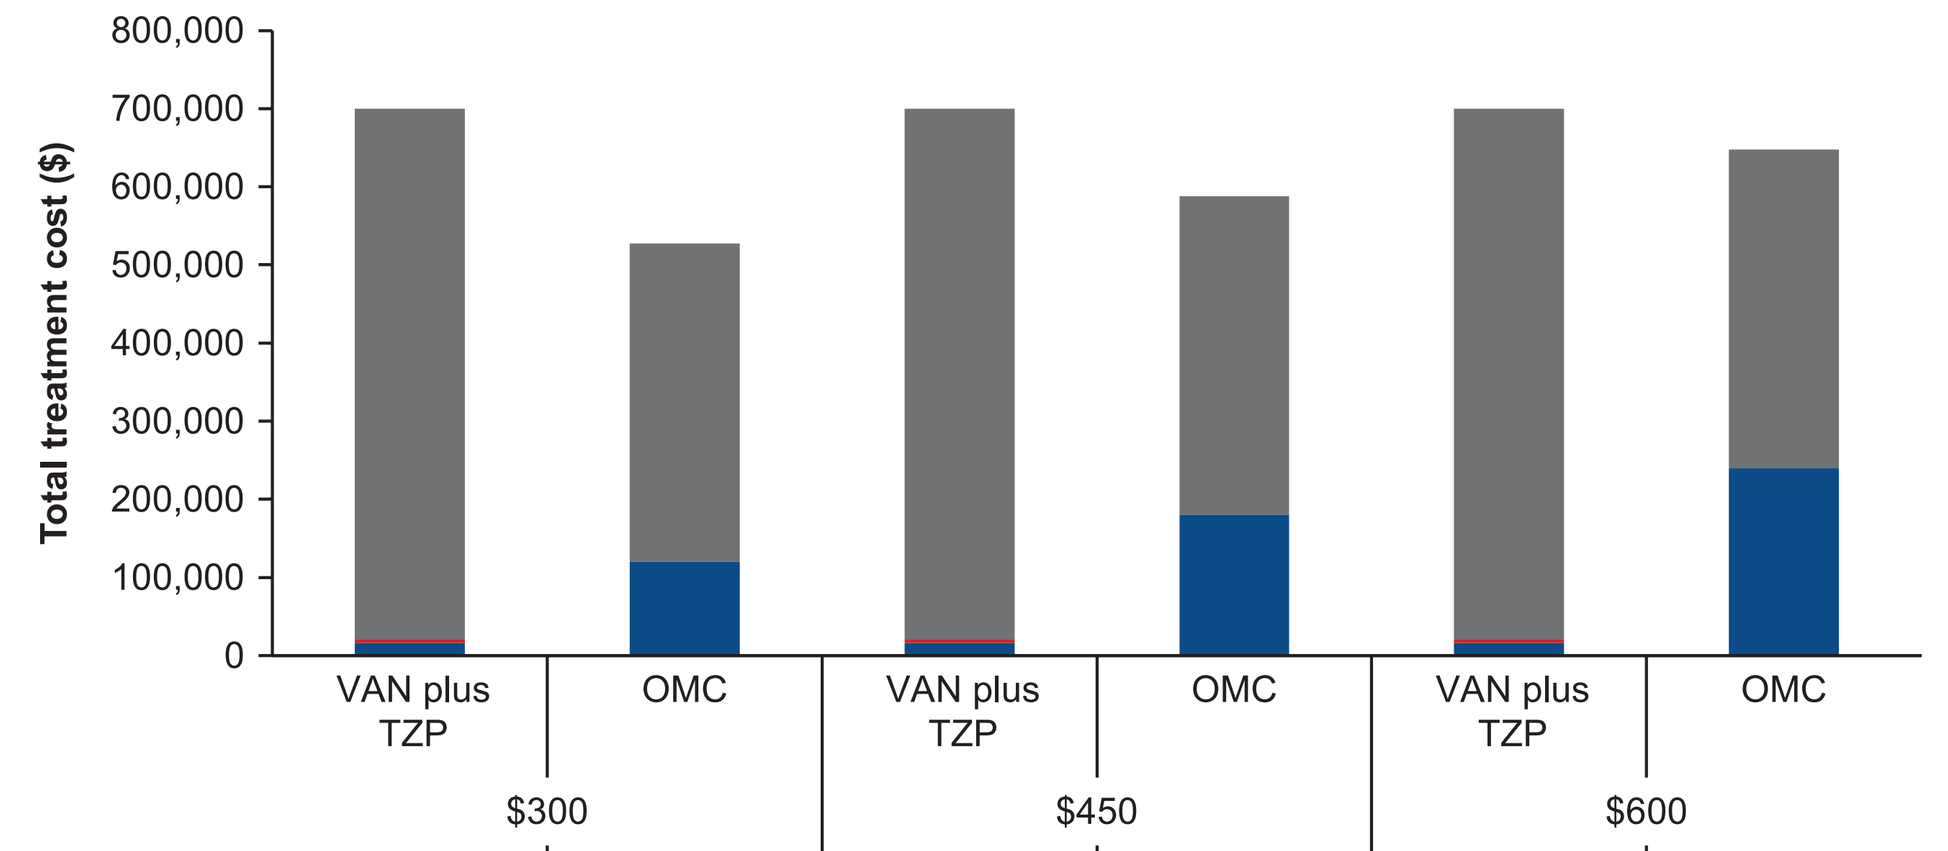
Total treatment cost ($)
800,000
700,000
600,000
500,000
400,000
300,000
200,000
100,000
0

	$300 VAN plus TZP	$300 OMC	$450 VAN plus TZP	$450 OMC	$600 VAN plus TZP	$600 OMC
Inpatient costs	679,419	407,651	679,419	407,651	679,419	407,651
Monitoring cost	4,362	0	4,362	0	4,362	0
Drug acquisition cost	16,170	120,000	16,170	180,000	16,170	240,000

OMC acquisition cost per day ($)

Abbreviations: LOS, length of stay; OMC, omadacycline; TZP, piperacillin-tazobactam; VAN, vancomycin.

DISCUSSION

- In line with previous studies,[4,5] the analysis assumes that patients with inadequate early clinical response can potentially be discharged 1 or 2 days earlier with OMC relative to SOC treatment because antibiotics like OMC with IV and oral bioequivalent formulations can safely and effectively reduce LOS
- The economic model is consistent with previous research demonstrating IV-PO antibiotics reduce hospitalization LOS[4] and suggest that targeted prescribing of OMC for treatment of ABSSSI patients with an inadequate early clinical response could result in cost-savings relative to the current SOC
- The model employed in this study shows that the use of OMC at price points above that of VAN plus TZP can be cost-minimizing compared with SOC through the reduction of hospital-associated services
- OMC and other treatments, which provide an option of IV to a bioequivalent oral formulation, offer healthcare systems a potential opportunity to promote efficient healthcare resource use. Efficient use of resources can likewise help providers meet the needs of payers as they migrate toward value-based payment models. The modeling supports this by showing that as LOS drops, OMC is cost-minimizing at acquisition costs above that of SOC
- Antibiotic stewardship is supported by shortening the duration of antibiotic use in the hospital setting along with LOS, and by the shifting of polypharmacy regimens to monotherapy,[11] both of which could be achieved with OMC use as modeled. The concept of high quality of care at lowest cost is congruent with the goals of antibiotic stewardship programs, which have a dual responsibility to optimize clinical outcomes and minimize healthcare utilization and costs without adversely impacting quality of care
- The reduction in LOS also reduces the risk of patients developing other hospital-acquired infections[12]
- Minimizing duration of antibiotic use also has the potential to limit unintended consequences of antibiotic use, such as the emergence of antibiotic resistance and adverse drug events

CONCLUSIONS

- Early, targeted use of OMC has the potential to reduce the economic burden of hospitalized patients with ABSSSI who have an inadequate early response to VAN plus TZP through an average reduction of LOS among these patients by 1 to 2 days
- Modern antibiotic stewardship should consider appropriate clinical diagnosis techniques, therapy choice, and treatment duration to take full advantage of potential clinical and economic benefits of novel antimicrobial agents like OMC
- It would be valuable to assess the economic benefits of early, targeted use with OMC in a real-world setting

REFERENCES

1. Degener F, et al. *Value Health.* 2015;18(7):A580-A581.
2. LaPensee K, Fan W. 2012 ISPOR Annual Meeting; June 2-6, 2012; Washington, DC. Poster PIN22.
3. Amara S, et al. *Curr Med Res Opin.* 2013;29(7):869-877.
4. Nathwani D, et al. *Clin Microbiol Infect.* 2014;20(10):993-1000.
5. Nathwani D, et al. *Int J Antimicrob Agents.* 2016;48(2):127-136.
6. Corey GR, et al. *Clin Infect Dis.* 2010;51(6):641-650.
7. Medi-Span® Price-Rx® (online analysis tool). Hudson, OH: Wolters Kluwer Clinical Drug Information; 2016. www.wolterskluwerCDI.com.
8. Bureau of Labor Statistics. Consumer Price Index. 2016. https://www.bls.gov/cpi/.
9. Centers for Medicare and Medicaid Services. Premier Hospital Historical Data [database online]. 2016. https://www.healthdata.gov/dataset/premier-hospital-historical-data.
10. Centers for Medicare and Medicaid Services. *2016 Clinical Laboratory Fee Schedule.* Revised January 2016. https://www.cms.gov/Medicare/Medicare-Fee-for-Service-Payment/ClinicalLabFeeSched/Clinical-Laboratory-Fee-Schedule-Files.html.
11. Barlam TF, et al. *Clin Infect Dis.* 2016;62(10):e51-77.
12. World Health Organisation. Fact Sheet: Health care-associated infections. Geneva: World Health Organization; 2016. http://www.who.int/gpsc/country_work/gpsc_ccisc_fact_sheet_en.pdf.

ACKNOWLEDGMENTS

KP and TL received consulting payments from Paratek Pharmaceuticals Inc.

CG, HL, CM, RM, and KY are employees of PAREXEL International, a company commissioned by Paratek Pharmaceuticals, Inc., to perform the analyses.

Medical editorial assistance, funded by Paratek Pharmaceuticals, Inc., was provided by Healthcare Consultancy Group.

Sponsorship Statement: Model and poster development were funded by Paratek Pharmaceuticals, Inc.

Presented at: AMCP NEXUS 2017; October 16-19, 2017; Dallas, TX. Abstract L1.